P #334

Functional Validation of Single Domain Antibody-Derived CD33 Specific CAR-T Cells for the Treatment of Acute Myeloid Leukemia

Brikena Gjeci, Salar Khan, Julian Scherer, Tirtha Chakraborty

Vor Biopharma Inc., Cambridge, MA, USA

INTRODUCTION

- CAR T cells have exhibited efficacious treatment of hematological malignancies such as acute myeloid leukemia (AML).
- Conventional CAR-T cells are generated using single-chain variable fragments (scFv) derived from two domains (V_L and V_H) of monoclonal murine antibodies, potentially leading to misfolding and anti-CAR immunogenicity.
- Camelid-derived single domain antibodies (sdAbs) have high thermal stability and refolding capacity due to their reduced size. Single domain antibodies can access novel epitopes and exhibit reduced immunogenic potential [1], suggesting their suitability as binders in CARs.

OBJECTIVE

- Focusing on CARs targeting CD33, we here validate novel sdAb-derived CARs (sdCD33CAR) in a semi-automated multi-step process including cell-cell avidity and compare them to traditional scFv-derived CARs.

METHODS

- ~10^{11} vicuña sdAbs were panned against soluble and cell-associated CD33 to identify distinct sdAb binders by ELISA and FACS. Ten binders were selected, expressed as soluble human Fc tagged molecules, and screened for affinity by Octet. Five binders were used for sdCARs with 4-1BB costimulatory domains.
- Lentiviral vector (LVV) was generated with a liquid handler to express CARs in an IL-2 Reporter System (CAR-IRS cells). CAR-IRS cells enable functional CAR validation by activation-dependent induction of a fluorescent reporter.
- CAR-IRS activation was assessed by flow cytometry or continuous IncuCyte® live cell imaging. CAR-IRS cells were also tested for cell-cell avidity against CD33-expressing target cells using the Z-Movi® platform.

RESULTS

Fig. 1. Illustration of the IL-2 Reporter System

- **Step 1**: Stable integration of the IRS construct into the Jurkat cell genome, which contains FP1 (mTurquoise) under the constitutively active EF1α promoter, and FP2 (mOrange) under the NFAT-sensitive IL2 promoter.
- **Step 2**: After lentiviral transduction, CAR protein expression results in CAR-IRS cells.
- **Step 3**: Coculture of CAR-IRS cells with antigen-expressing cells induces antigen binding and CAR-IRS cell activation. NFAT production activates the IL2 promoter and results in quantifiable FP2 expression, which is used as proxy for CAR-induced cell activation.

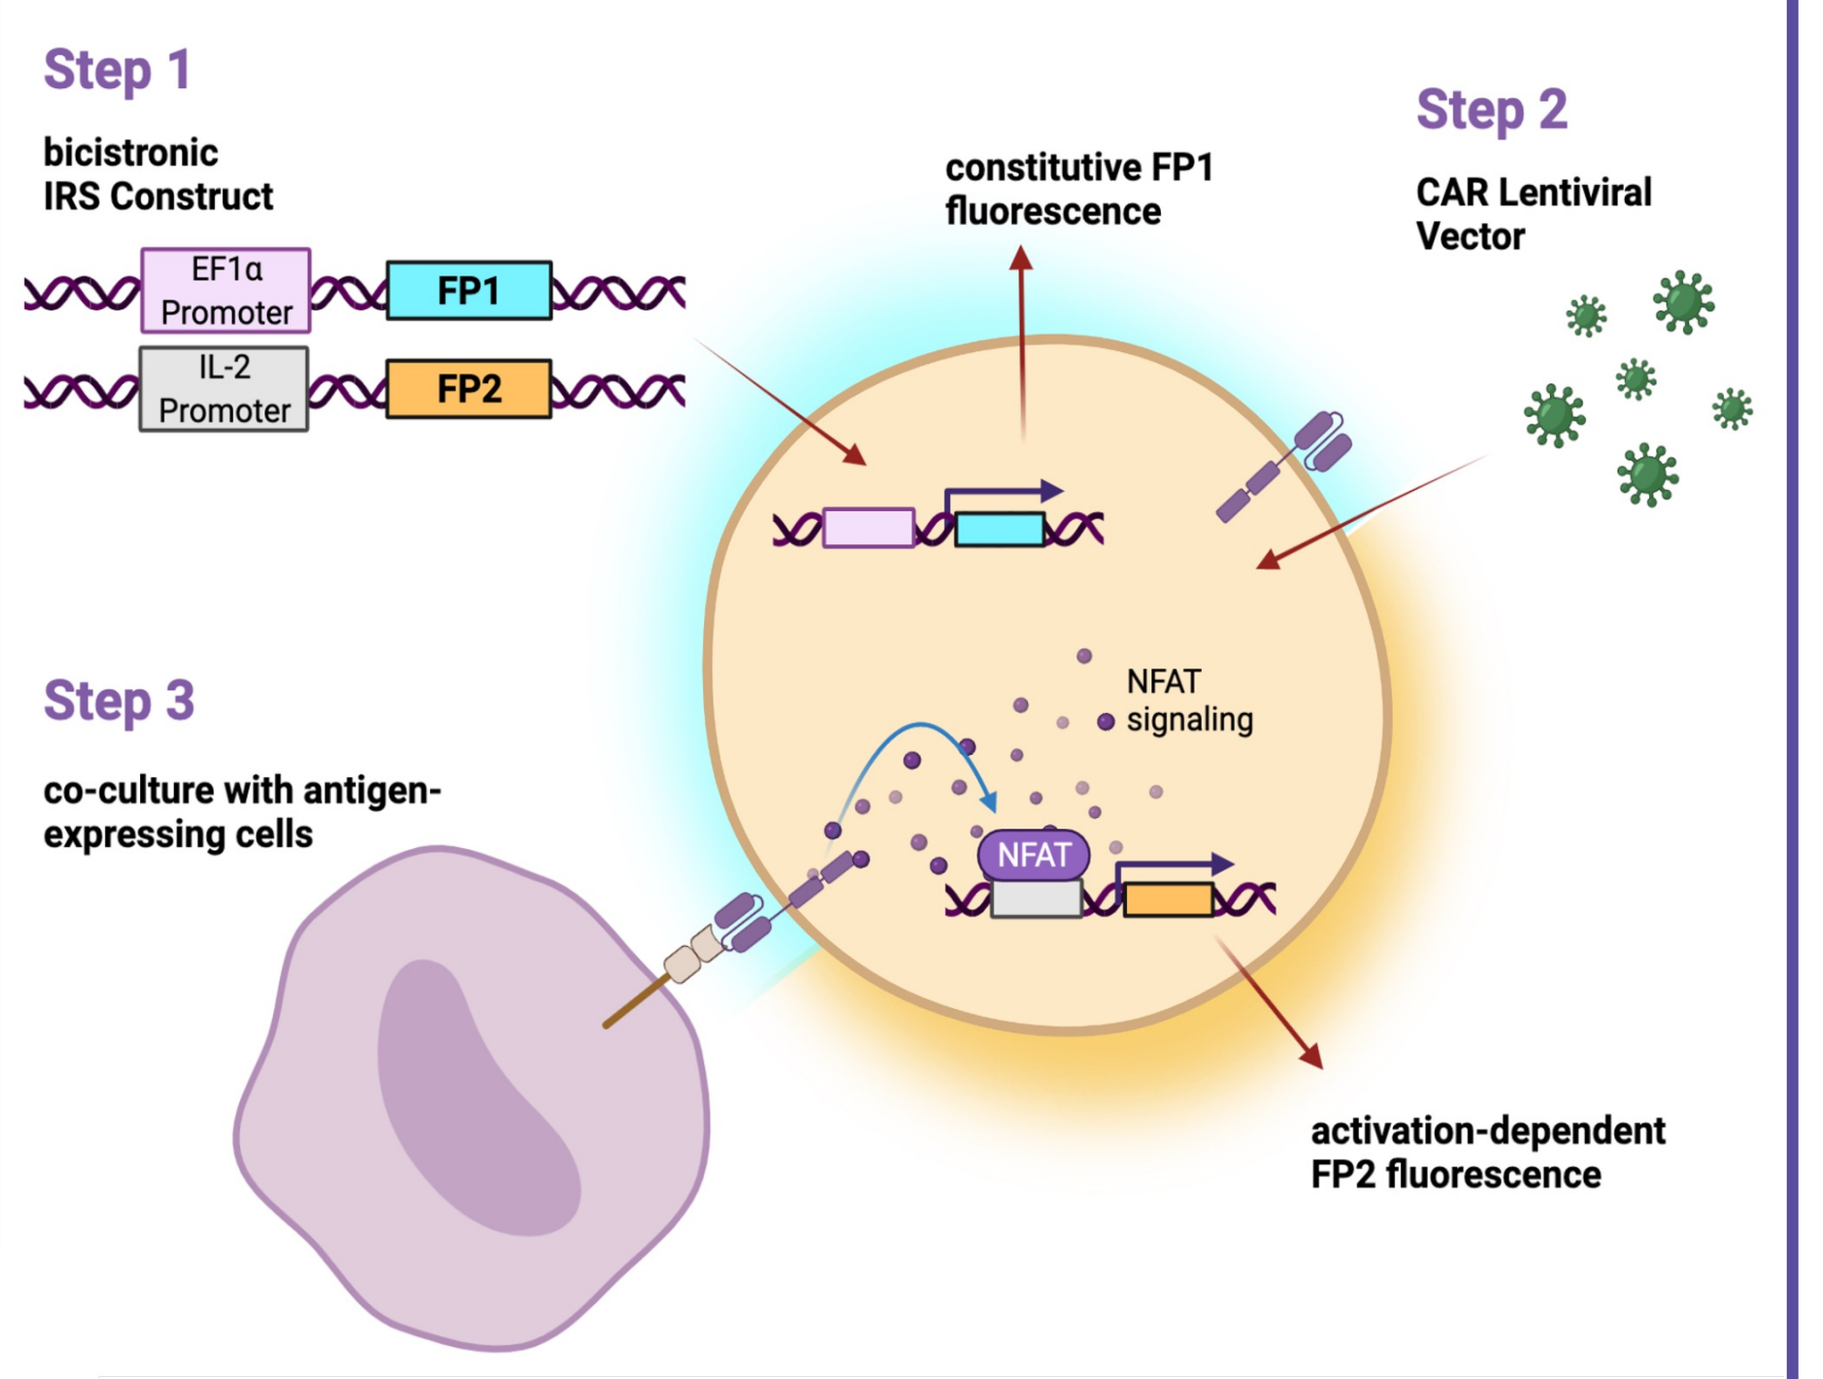

Step 1
bicistronic IRS Construct
EF1α Promoter
FP1
IL-2 Promoter
FP2
constitutive FP1 fluorescence
Step 2
CAR Lentiviral Vector
NFAT signaling
NFAT
Step 3
co-culture with antigen-expressing cells
activation-dependent FP2 fluorescence

RESULTS (CONT'D)

Fig. 2. CAR Structure and Screening Platform

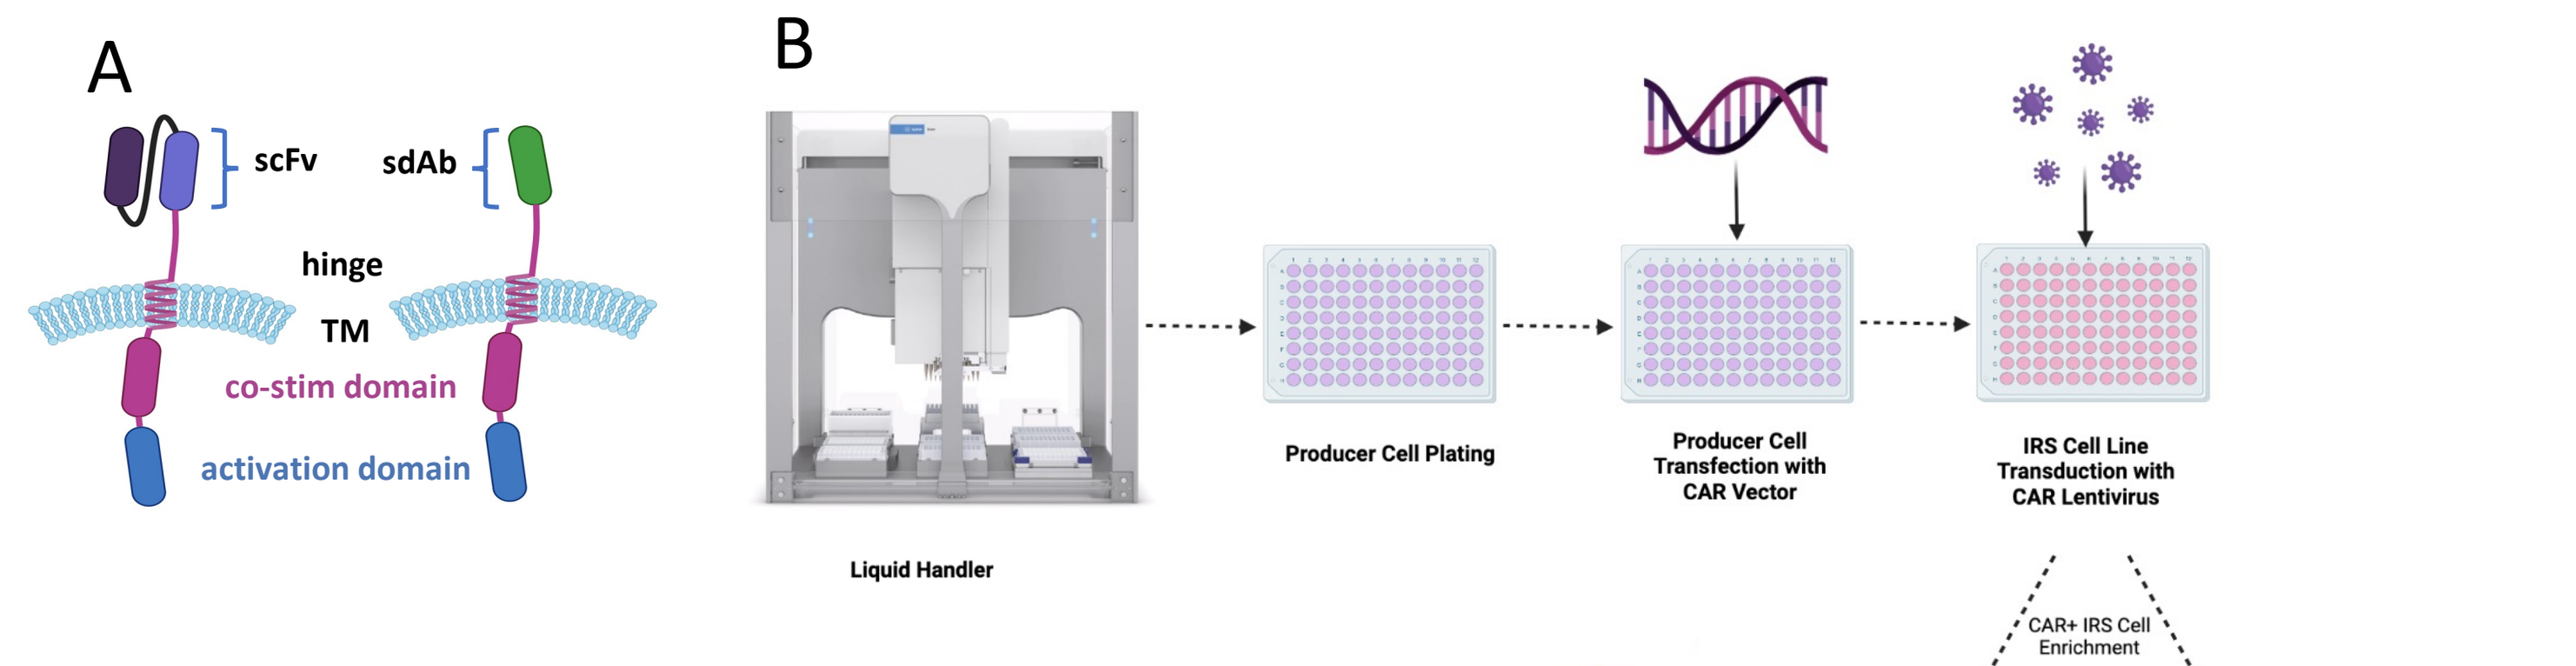

A
scFv
sdAb
hinge
TM
co-stim domain
activation domain
B
Liquid Handler
Producer Cell Plating
Producer Cell Transfection with CAR Vector
IRS Cell Line Transduction with CAR Lentivirus

- A) Schematic of the CAR structures for single chain variable fragment (scFv) and single-domain antibody (sdAb) binder classes. The antigen binding site of scFv CARs contain the variable regions of antibody heavy and light chains connected through a flexible linker. The sdAb CARs consist of a single heavy chain domain. Additional components (hinge, transmembrane region (TM), co-stimulation, and activation domains) of the CAR molecule are similar for both classes of binders.
- B) CAR screening is semi-automated and allows for 96well plate throughput: A liquid handler is utilized for lentiviral production and transduction of IL-2 reporter system (IRS) Jurkat cell lines. CAR positive IRS cells are enriched to >80% purity. Final read outs of the screen include antigen-specific CAR activity by live cell analysis and flow cytometry as well as CAR-IRS cell avidity with target cell lines.

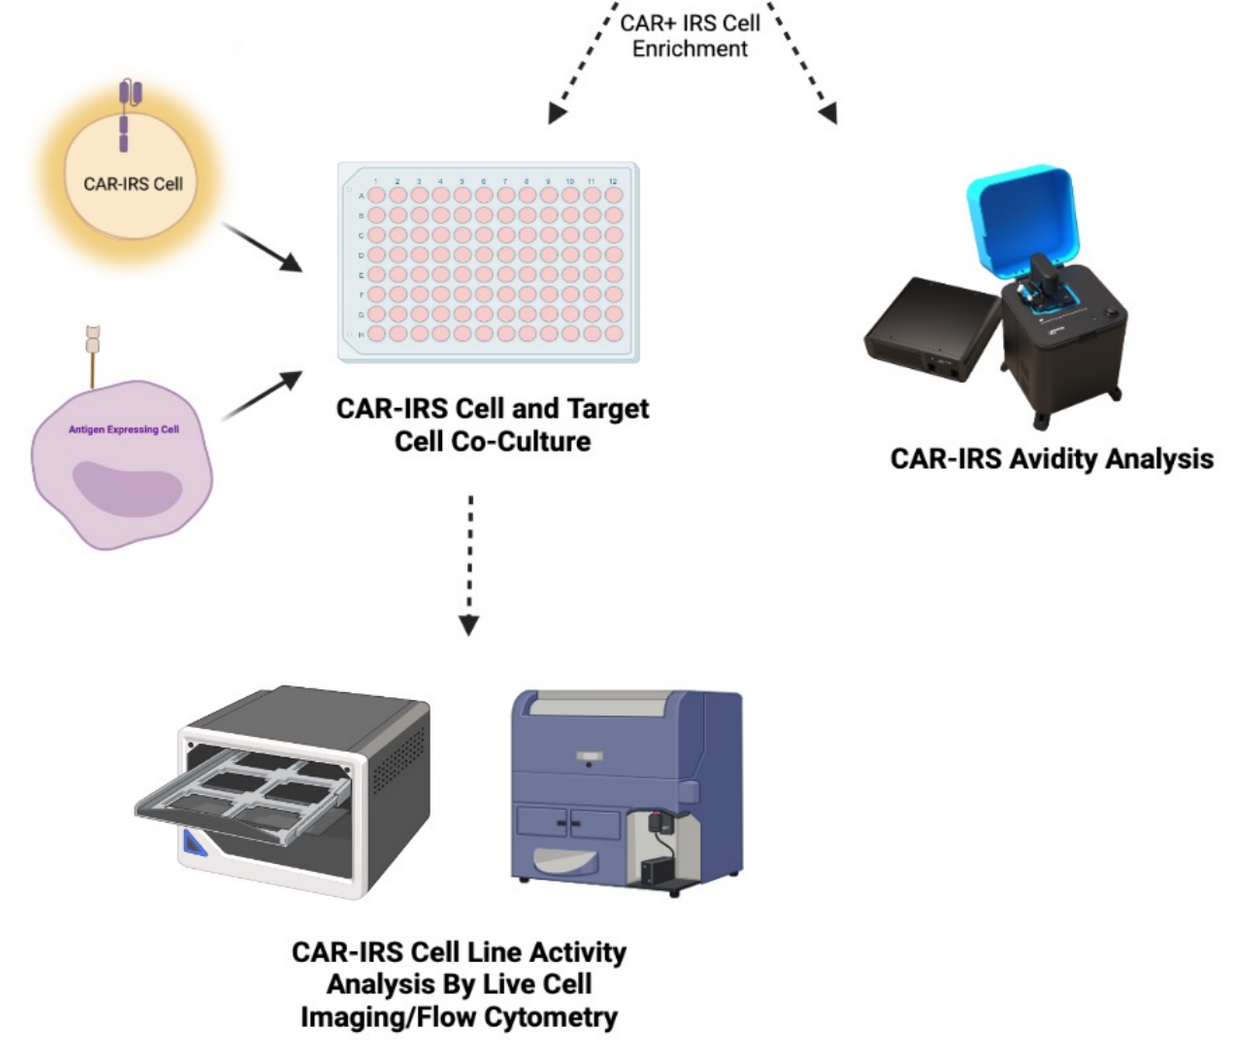

CAR+ IRS Cell Enrichment
CAR-IRS Cell
CAR-IRS Cell and Target Cell Co-Culture
CAR-IRS Avidity Analysis
CAR-IRS Cell Line Activity Analysis By Live Cell Imaging/Flow Cytometry

Fig. 3. List of Tested CD33-directed CAR Constructs

CAR #	Binder Name	Binder Class	Linker	TM	Co-Stim	Activation
1	Vor Binder 1	sdAb	CD8a	CD8a	4-1BB	CD3z
2	Vor Binder 2	sdAb	CD8a	CD8a	4-1BB	CD3z
3	Vor Binder 3	sdAb	CD8a	CD8a	4-1BB	CD3z
4	Vor Binder 4	sdAb	CD8a	CD8a	4-1BB	CD3z
5	Vor Binder 5	sdAb	CD8a	CD8a	4-1BB	CD3z
6	Lintuzumab (huM195)	scFv	CD8a	CD8a	4-1BB	CD3z
7	Gemtuzumab (hP67.6)	scFv	CD28	CD28	CD28	CD3z
8	My9-6	scFv	CD8a	CD8a	4-1BB	CD3z

A selection of eight CAR constructs were enrolled in the screen to compare newly identified sdAb binders with scFv binders. Binder class, binder name, linker, transmembrane region (TM), and intracellular co-stimulation and activation domains of each tested CAR construct are listed.

Fig. 4. Single-Domain Antibody Binders Show Affinities in the Nanomolar Range Comparable to scFv

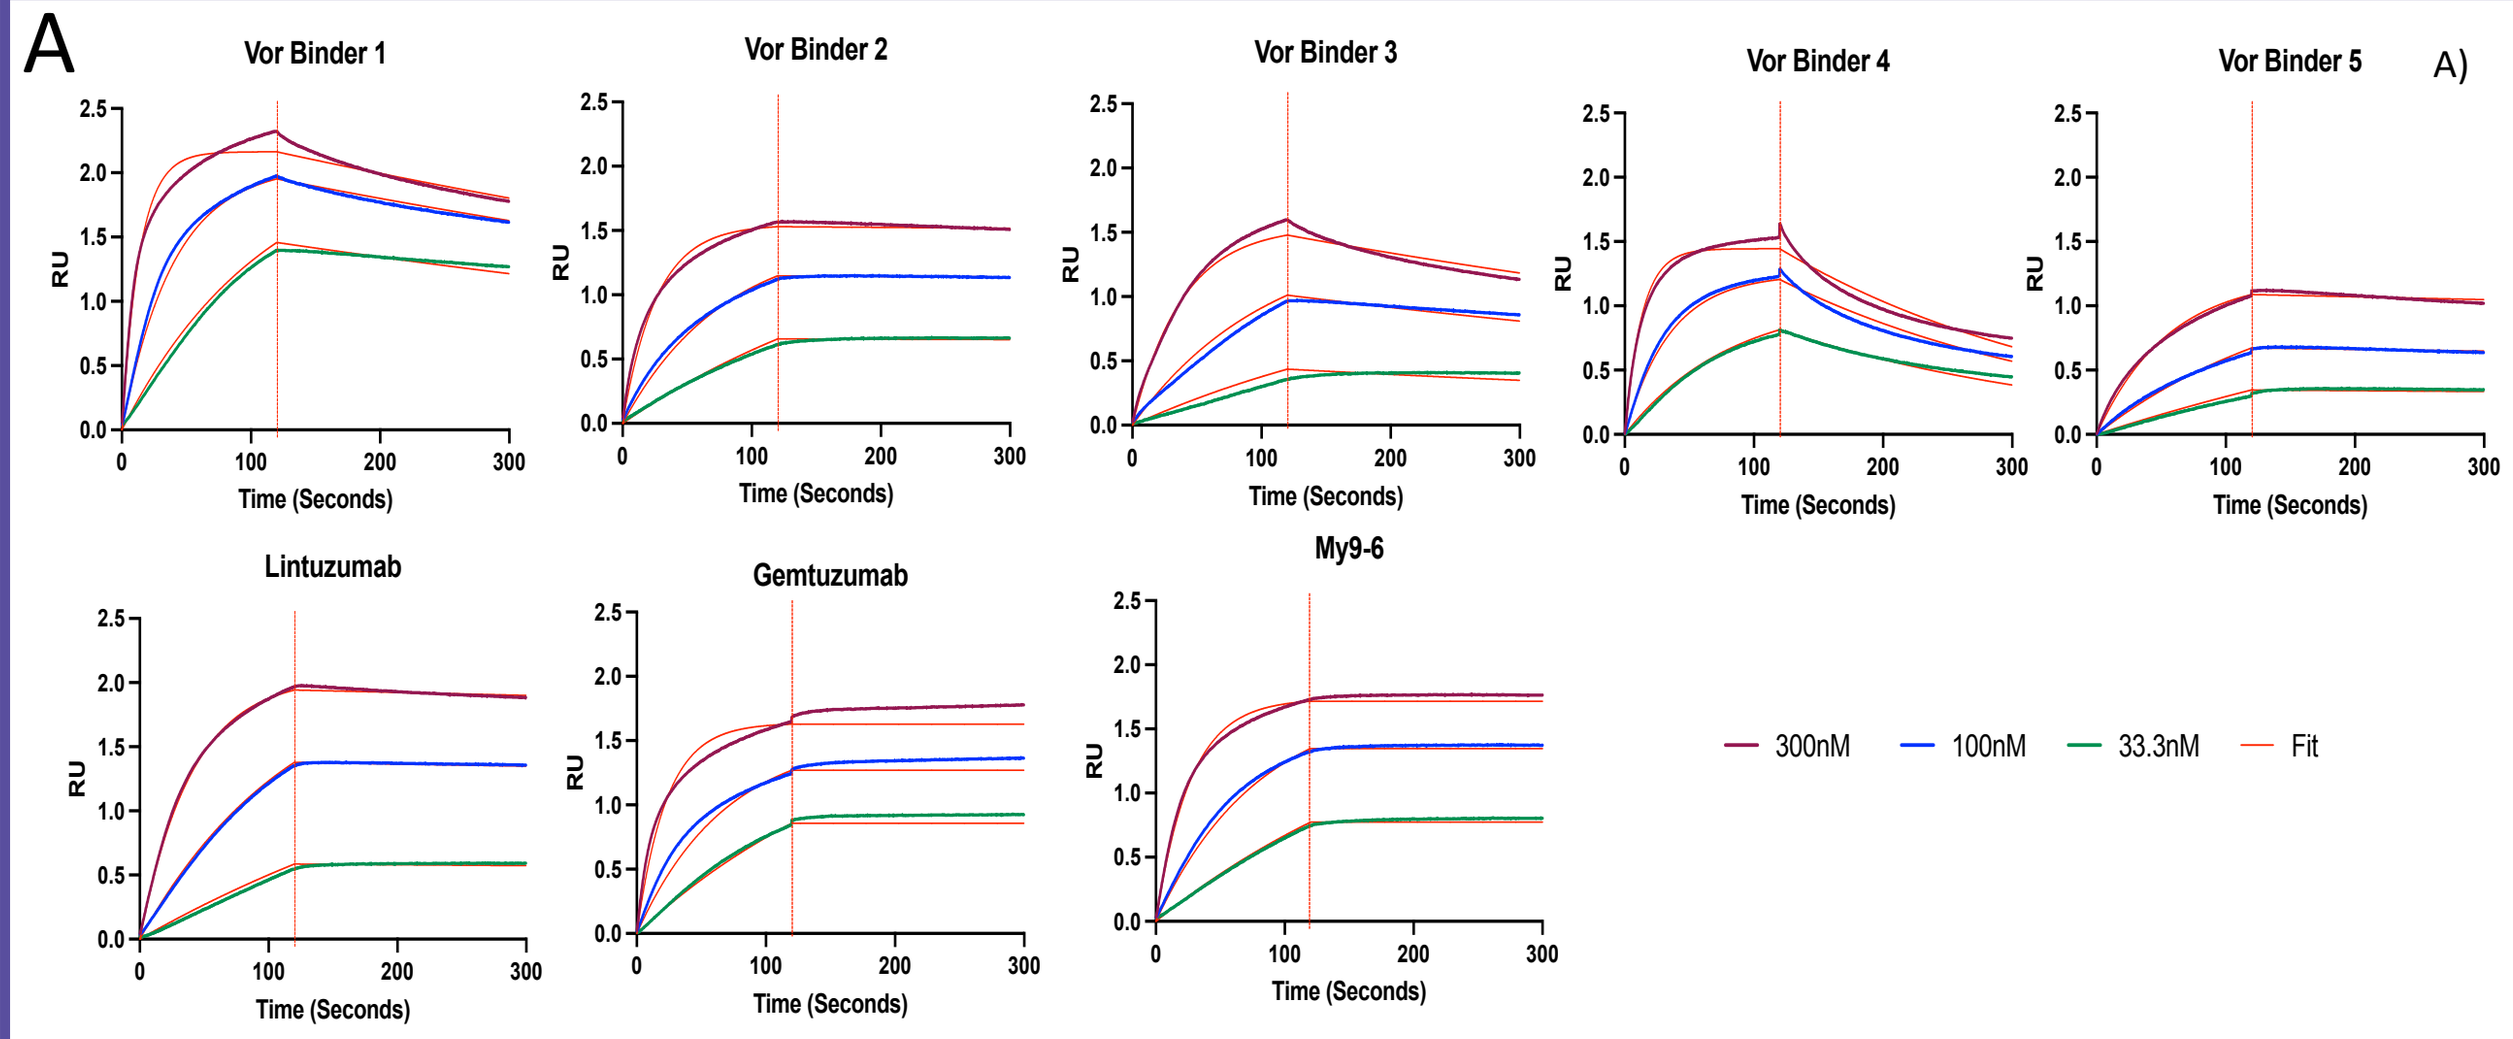

A
Vor Binder 1
Vor Binder 2
Vor Binder 3
Vor Binder 4
Vor Binder 5
Lintuzumab
Gemtuzumab
My9-6
RU
Time (Seconds)
300nM
100nM
33.3nM
Fit

B

	sdAb					IgG		
Sample ID	Vor Binder 1	Vor Binder 2	Vor Binder 3	Vor Binder 4	Vor Binder 5	Lintuzumab	Gemtuzumab	My9-6
kD (nM)	4.88	9.48	13.4	11.5	2.28	1.46	0.073 [2]	0.102 [3]
kon(1/Ms)	5.60E+05	2.03E+05	1.34E+05	3.64E+05	8.99E+04	8.31E+04	2.13E+05	1.23E+05
kdis(1/s)	2.73E-03	1.92E-03	1.79E-03	4.17E-03	2.05E-04	1.21E-04	ND	ND

A) Octet Binding curves of the scFv and sdAb binders against recombinant human CD33. Biotinylated human CD33 extracellular domain (ECD) was captured at 10µg/ml on streptavidin (SA) sensors. Analytes (scFv or sdAb binders) were kept in solution. Starting with a concentration of 300nM of analyte, 1:3 serial dilutions of the analytes were tested and the data points for each of the binders is shown in purple (300nM), blue (100nM) or green (33.3nM). The data fit for the binders and the dilutions is shown in red in all the plots. RU: response units.

B) Binding affinities (kD) and kinetics (on-rates, kon; off-rates, kdis) of the binders are shown in table format. ND: none detected.

Fig. 5. Co-Cultures Indicate Comparable Induction of T cell Activation for both Classes of CAR Binders

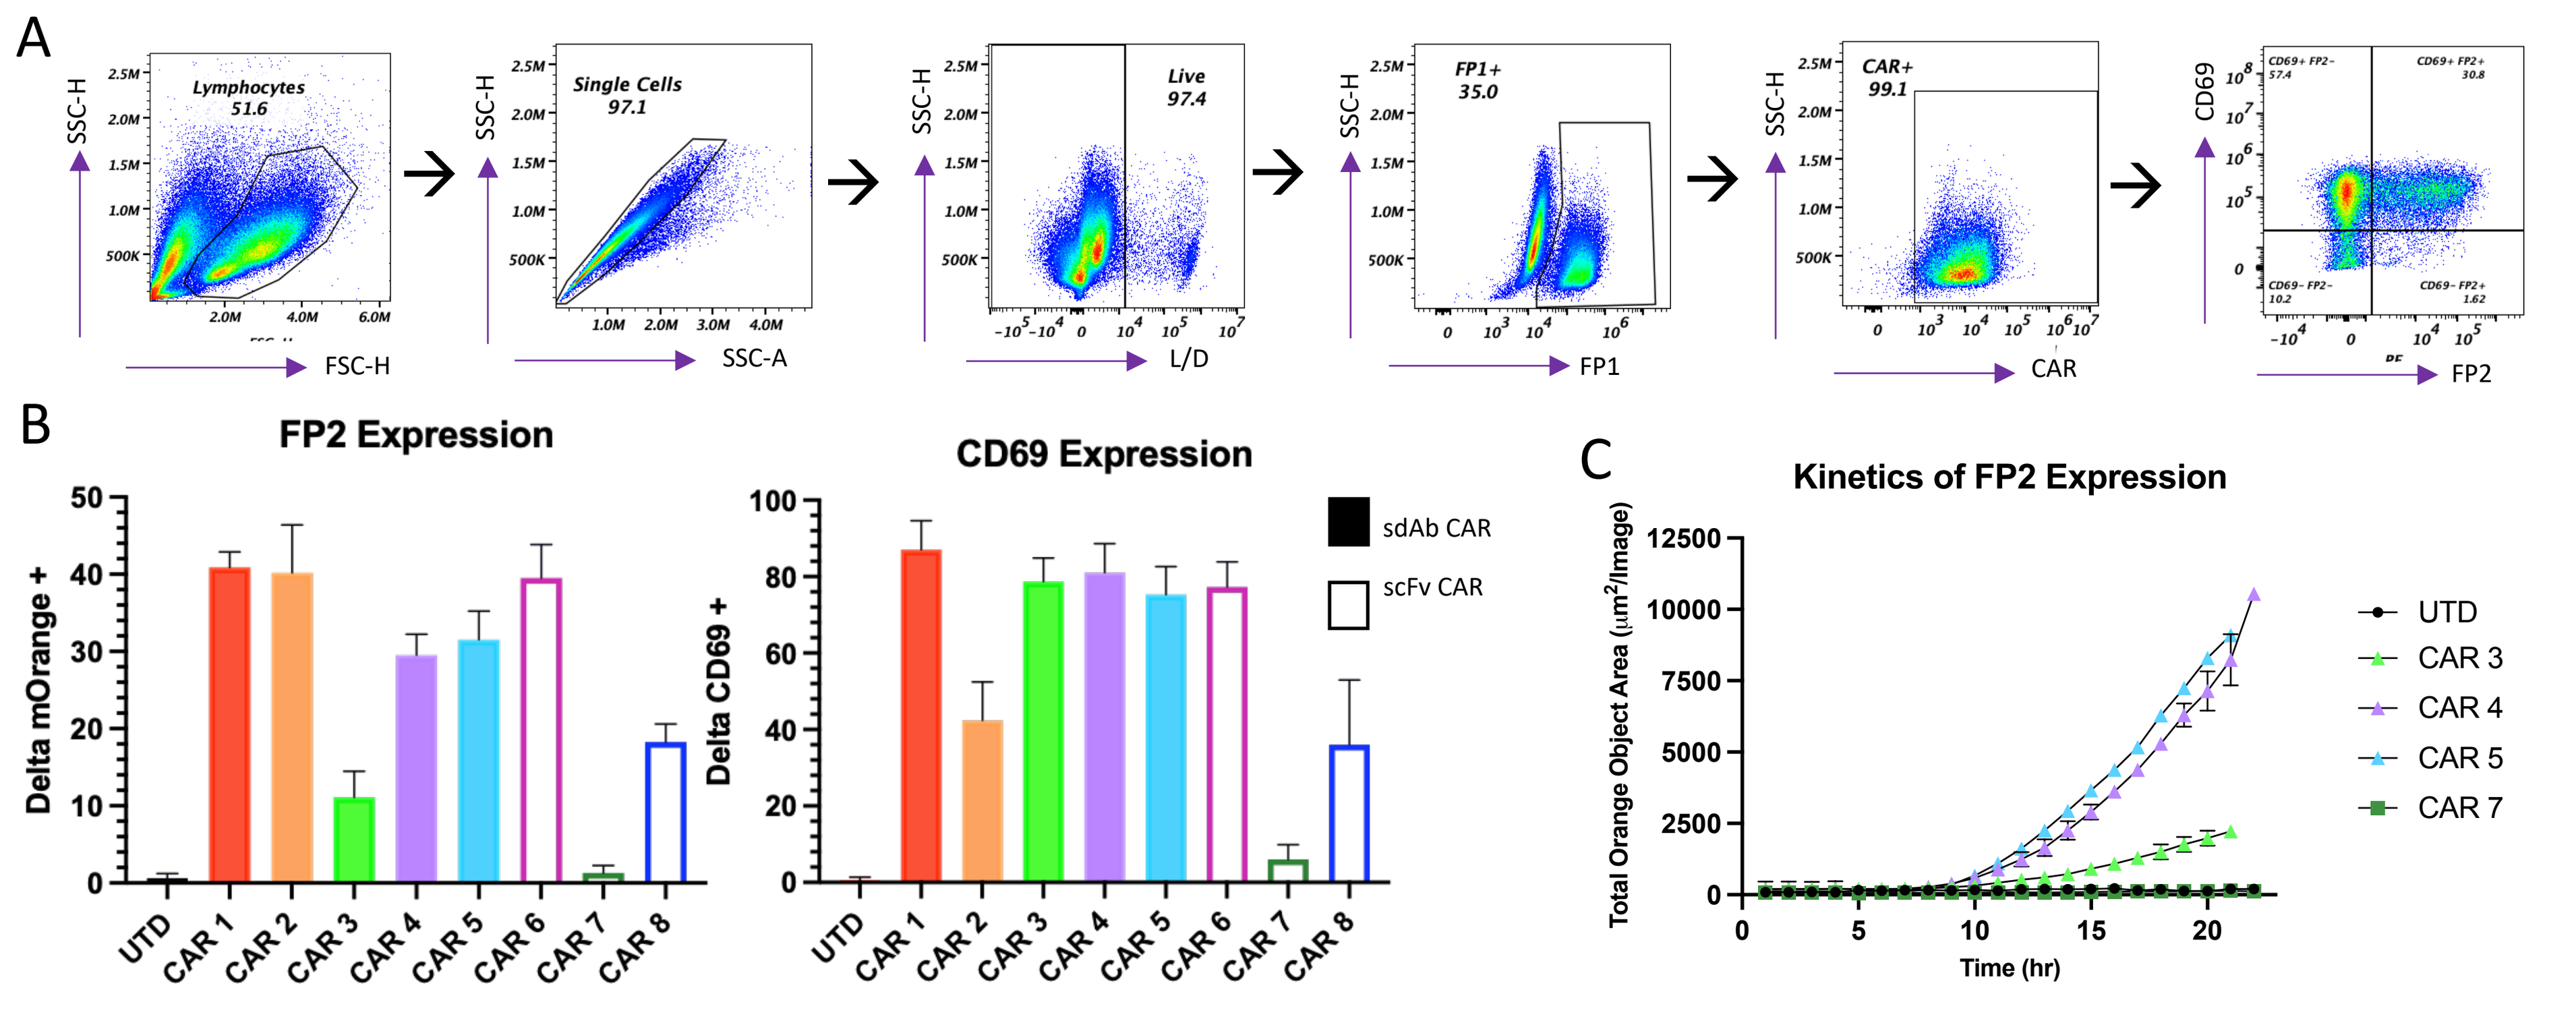

A
Lymphocytes 51.6
Single Cells 97.1
Live 97.4
FP1+ 35.0
CAR+ 99.1
SSC-H
FSC-H
SSC-A
L/D
FP1
CAR
CD69
FP2
B
FP2 Expression
CD69 Expression
Delta mOrange +
Delta CD69 +
sdAb CAR
scFv CAR
UTD
CAR 1
CAR 2
CAR 3
CAR 4
CAR 5
CAR 6
CAR 7
CAR 8
C
Kinetics of FP2 Expression
Total Orange Object Area (µm²/Image)
Time (hr)

A) Representative gating strategy to detect FP2 and CD69 signal on CAR-IRS cells after 24hr co-cultures with CD33-expressing MOLM-13 WT target cells. The same gating strategy was followed after MOLM-13 CD33KO co-cultures.

B) Bar graphs of FP2 and CD69 signal on CAR-IRS cells in response to MOLM13 target cell co-cultures. Results indicate the background subtracted difference (delta) in FP2 or CD69 percentage between WT and CD33KO co-cultures.

C) Kinetic profile of FP2 signal on select CAR-IRS cell lines during co-cultures with MOLM-13 WT cells using IncuCyte® live cell imaging. Profiles demonstrate CAR-IRS activation irrespective of binder format beginning at ~10 hr after the initiation of the co-cultures. Note: Rank order of selected CARs is consistent between IncuCyte and Flow Cytometric analysis.

Fig. 6. Process of CAR-IRS Avidity Measurements Utilizing the Z-Movi® Platform

- **Step 1**: CD33 expressing target cells (e.g.: MOLM-13 AML cells) are seeded onto avidity chip and allowed to adhere.
- **Step 2**: CAR-IRS Cells are fluorescently labeled and incubated with the target monolayer.
- **Step 3**: A force ramp is applied, inducing the detachment of CAR-IRS cells from the target cell layer revealing cell-cell binding strength and avidity.

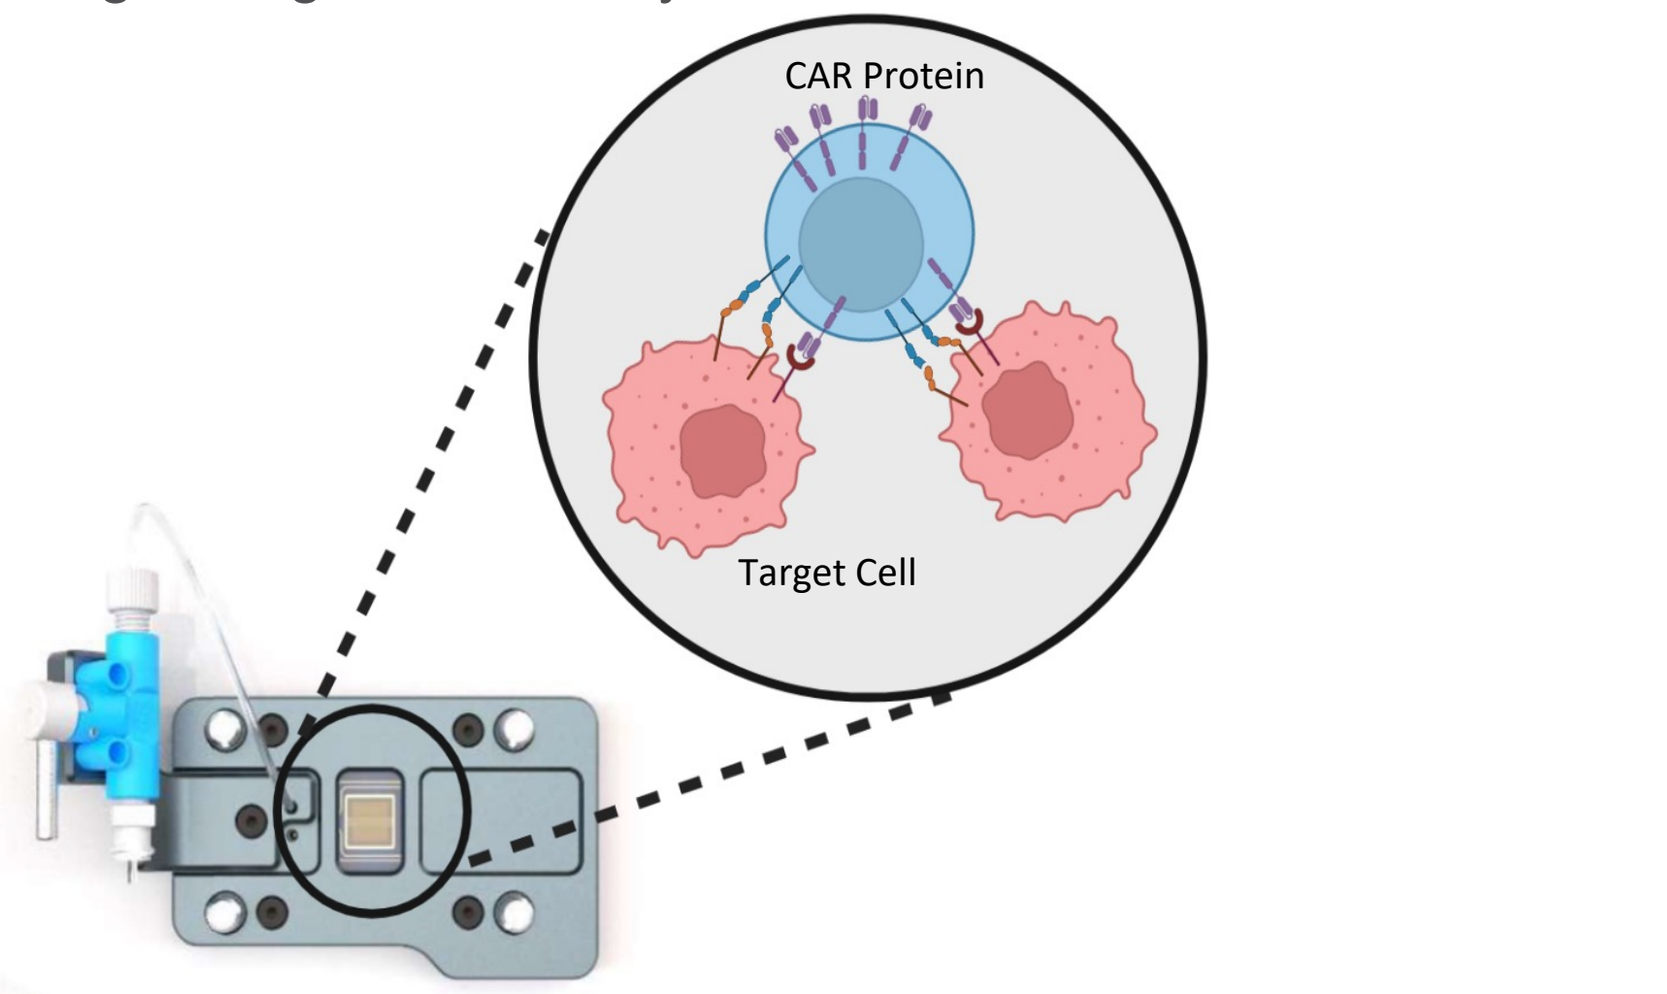

CAR Protein
Target Cell

Fig. 7. Z-Movi® Indicates Similar Avidity for Both Binder Classes

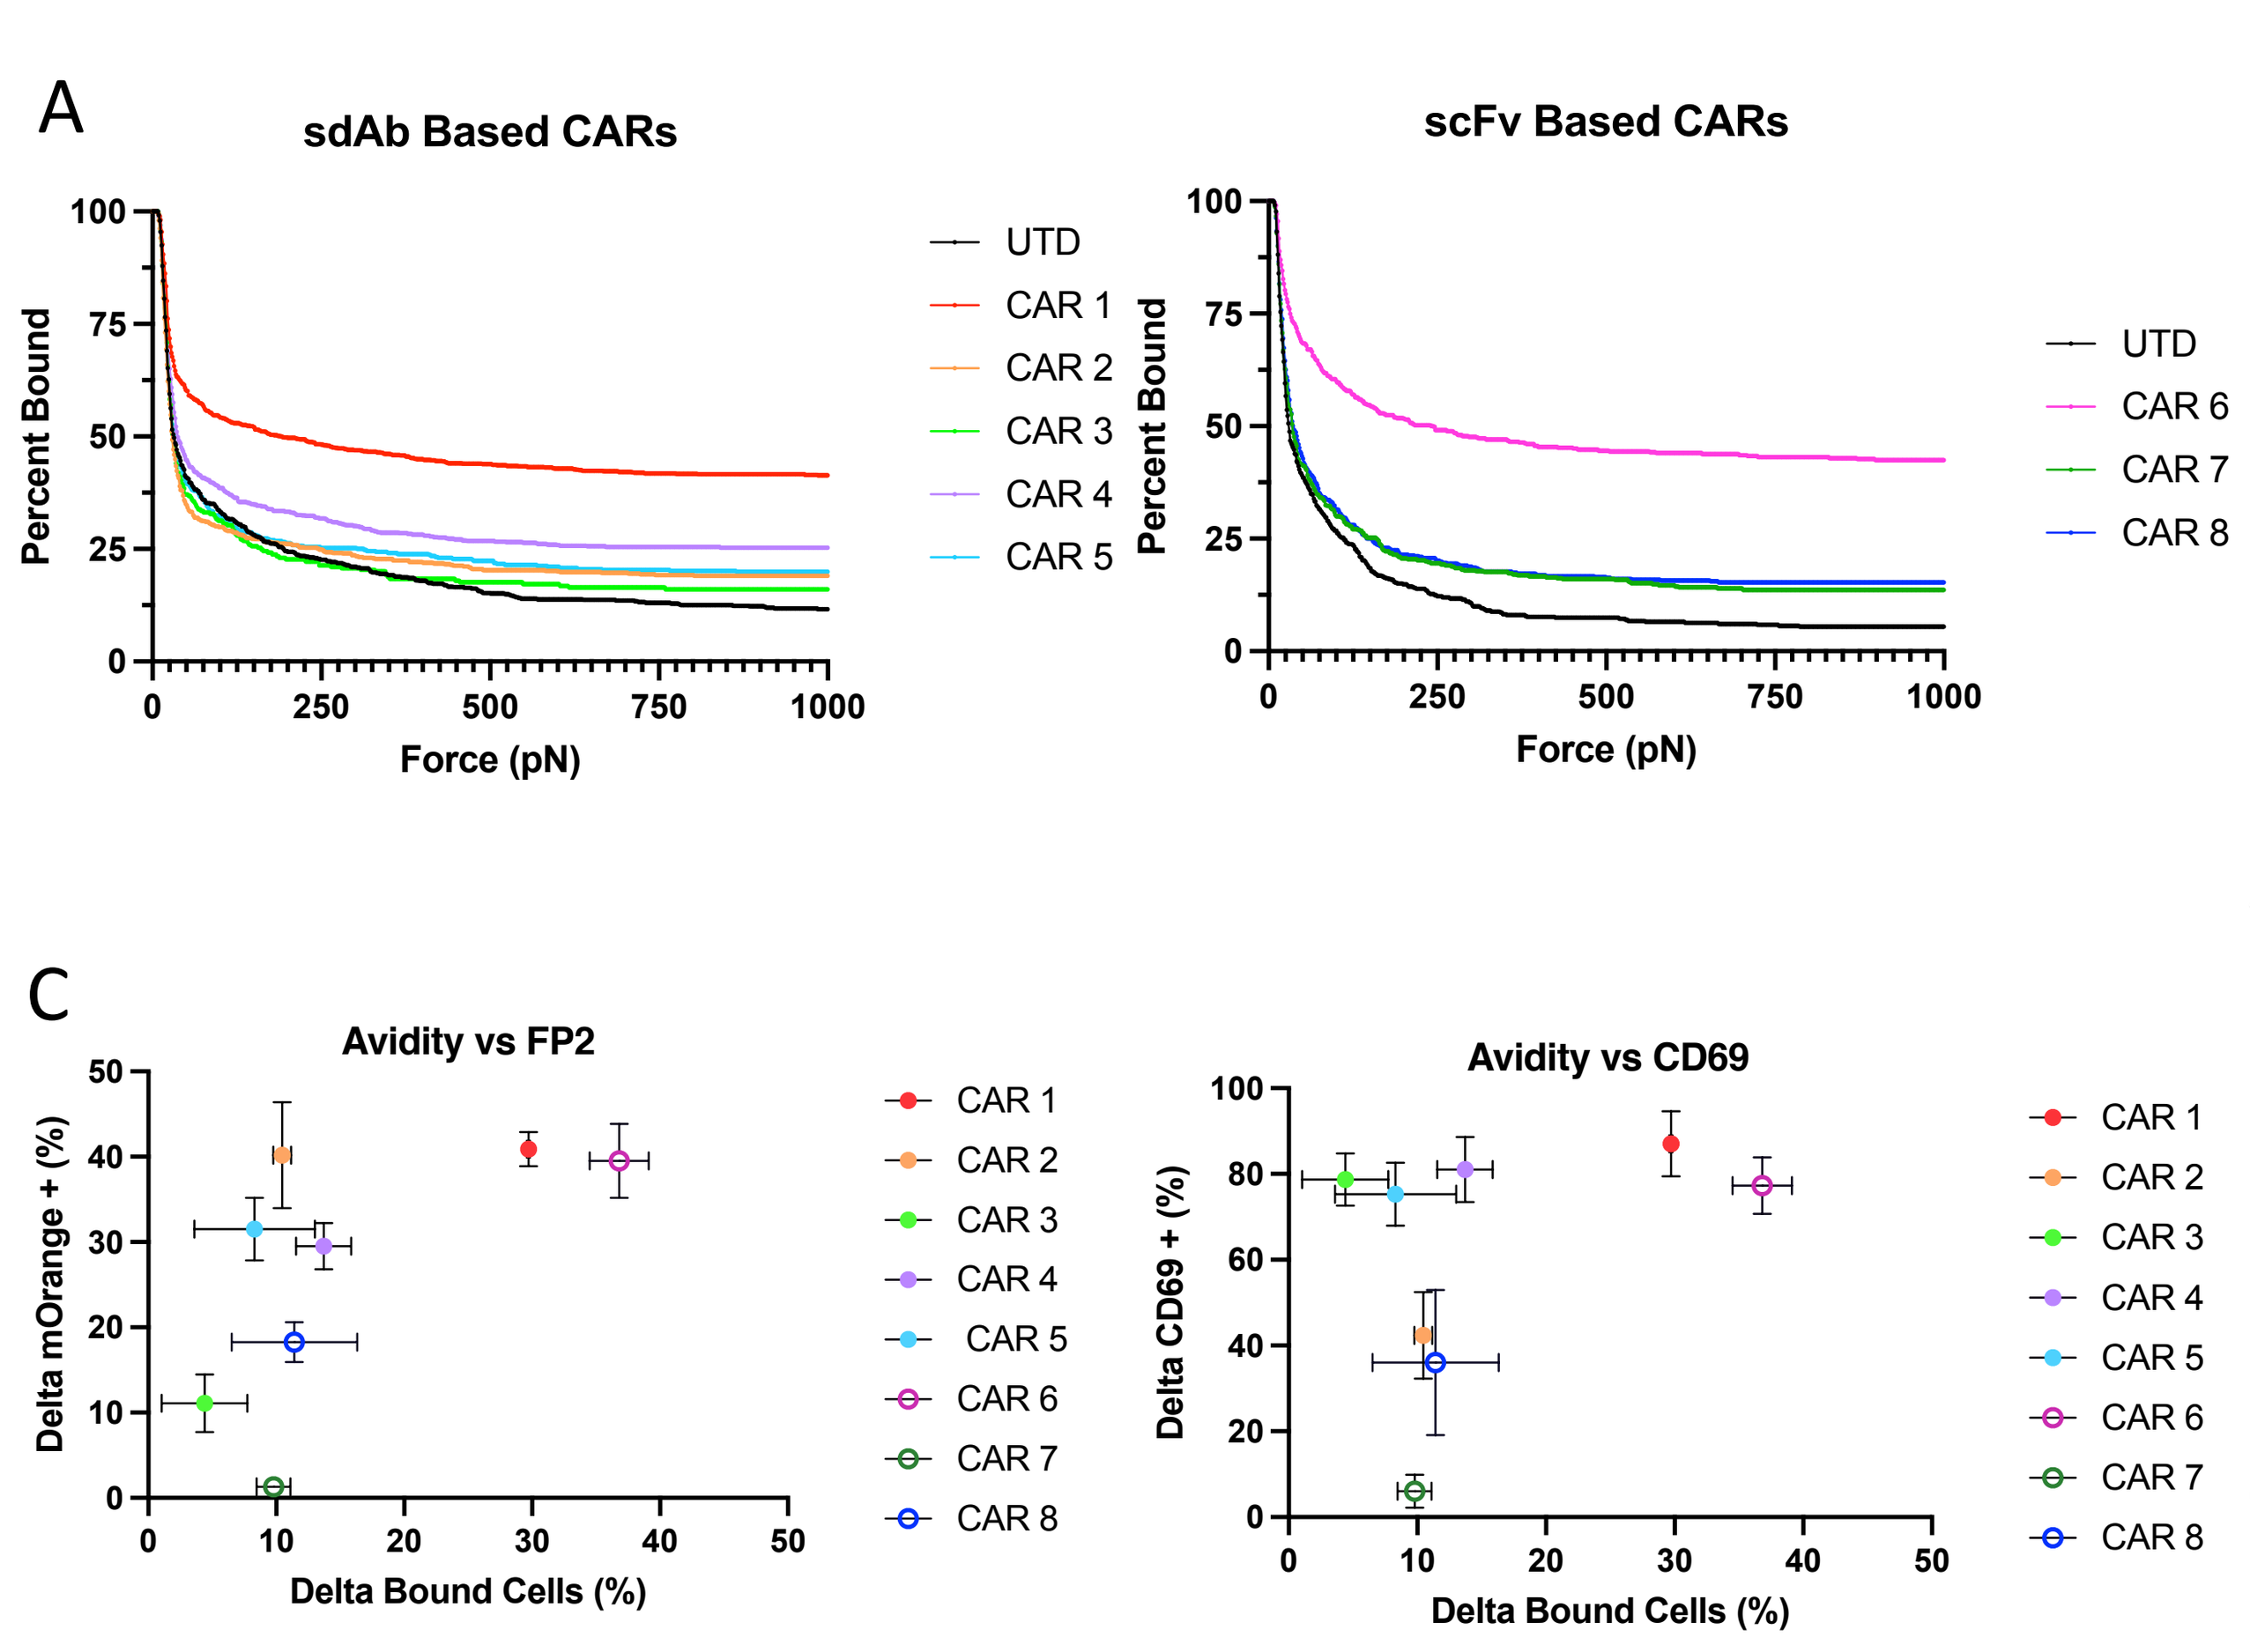

A
sdAb Based CARs
scFv Based CARs
Percent Bound
Force (pN)
C
Avidity vs FP2
Avidity vs CD69
Delta mOrange + (%)
Delta CD69 + (%)
Delta Bound Cells (%)

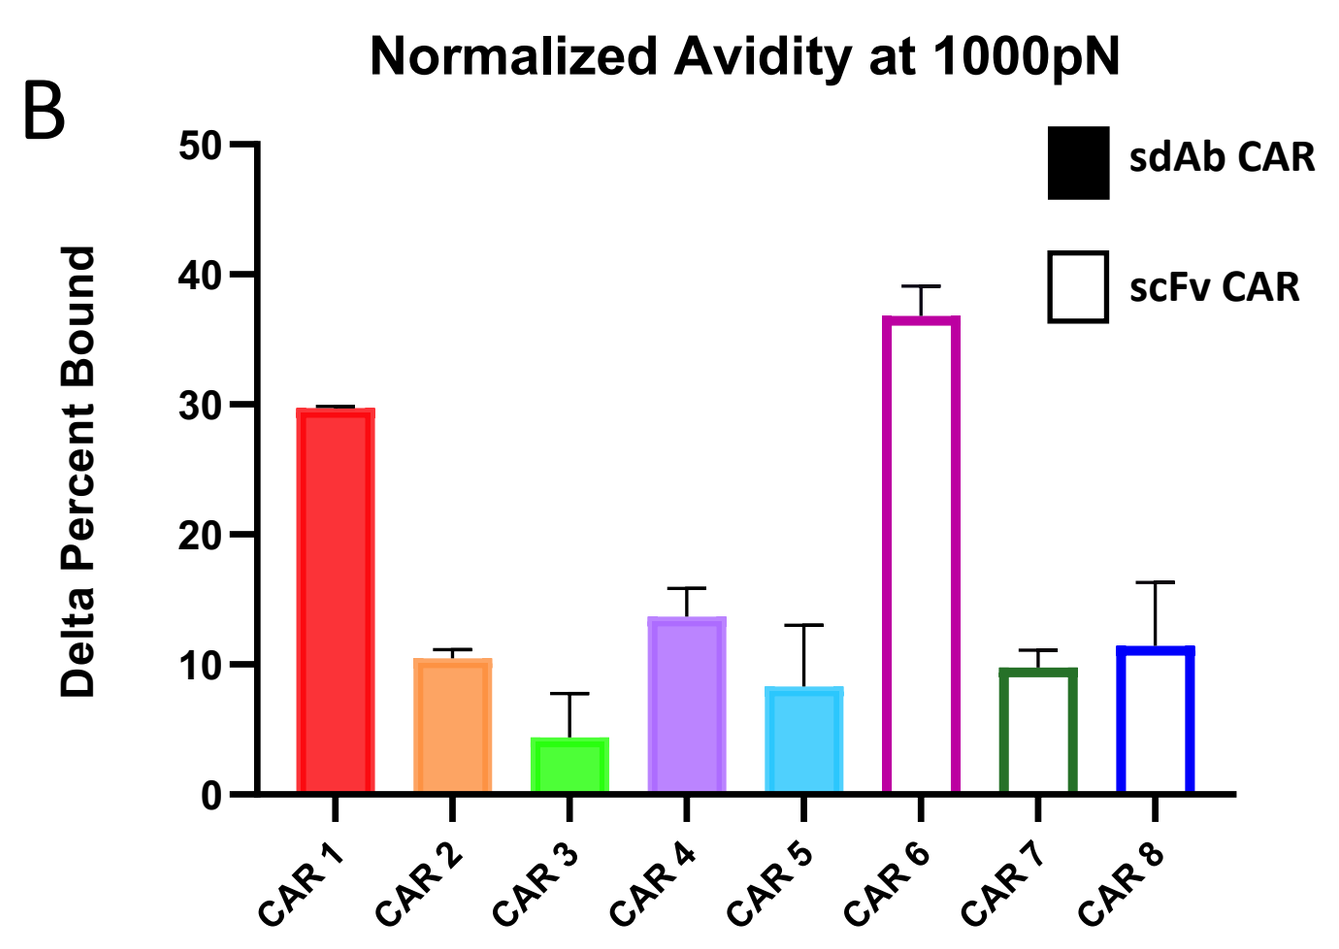

B
Normalized Avidity at 1000pN
Delta Percent Bound
sdAb CAR
scFv CAR

A) CAR-IRS cells were incubated inside the avidity chip with MOLM-13 WT cells and a force ramp (0-1000pN) was applied. Trajectories indicate the percent of CAR-IRS cells bound to target monolayer across the force ramp. UTD represents the untransduced IRS cell negative control. Single-domain antibody-based CARs on top, scFv-based CARs on the bottom.

B) Bar graph showing the normalized avidity value (delta percent bound) at 1000pN. Normalization was calculated by subtracting the UTD value. The top sdAb and scFv CARs have similar normalized avidity.

C) Comparison of avidity at 1000pN and activation data (either by FP2, left, or by CD69 signal, right) indicates limited correlation between the two parameters. CAR1 (sdAb) and CAR6 (scFv) were identified as potential lead anti-CD33 CAR constructs at the conclusion of the screen.

CONCLUSION

- Automated liquid handling allows for high-throughput lentiviral vector (LVV) transduction to generate CAR-IRS Jurkat cells, which enables reliable, fast, and economical screening across CAR constructs.
- Screening of CD33-directed CAR constructs included measurements of activation potential after co-culture with AML target cells and cell-cell avidity.
- The screen identified one single-domain antibody-based CAR construct, which shows results similar to traditional single-chain variable fragment (scFv)-derived CARs.
- These results provide a promising candidate for future *in vitro* and *in vivo* screening of primary CAR-T cell therapies against AML.

References

1. Asaadi Y, Jouneghani FF, Janani S, Rahbarizadeh F. A comprehensive comparison between camelid nanobodies and single chain variable fragments. Biomark Res. 2021 Dec 4;9(1):87. PMID: 34863296.
2. Ali S, Dunmore H, Karres D, Hay JL, Salmonsson T, Gisselbrecht C, Sarac SB, Bjerrum OW, Hovgaard D, Barbachano Y, Nagercoil N, Pignatti F. The EMA Review of Mylotarg (Gemtuzumab Ozogamicin) for the Treatment of Acute Myeloid Leukemia. The Oncolgist. 2019 May;24(5):171-9.
3. Hoseini SS, Vadlamudi M, Espinosa-Cotton M, Tran H, Feng Y, Guo H, Cheung I, Cheung NV. T cell engaging bispecific antibodies targeting CD33 IgV and IgC domains for the treatment of acute myeloid leukemia. JITC. 2021;9:e002509.

Acknowledgments

We thank the Research, Technical Operations, and Lab Operations teams at Vor Biopharma. Figures 1, 2 and 6 were generated in part with BioRender. Liquid Handler image in Figure 2 was obtained from: Agilent.com. Z-Movi image in Figure 2 was obtained from Lumicks.com

Presented at

SITC 2022, November 8–12, 2022.