Initial First-In-Human Results: CD33-Deleted Hematopoietic Stem and Progenitor Cells Display Normal Engraftment after Hematopoietic Cell Transplant (HCT) and Tolerate Post-HCT Gemtuzumab Ozogamicin (GO) without Cytopenias

LBA14

Benjamin Tomlinson,[1] Hyung C. Suh,[2] Divya Koura,[3] Guenther Koehne,[4] Christina Cho,[2] Nadia M. Bambace,[4] Léa Bernard,[5] Nirali N. Shah,[6] Roland B. Walter,[7] John F. DiPersio,[8] Miguel-Angel Perales,[9] Michael Loken,[10] Sritama Nath,[11] Glen D. Raffel,[11] Brenda W. Cooper[1]

[1]Adult Hematologic Malignancies & Stem Cell Transplant Section, Seidman Cancer Center, University Hospitals Cleveland Medical Center, Cleveland, OH, USA; [2]John Theurer Cancer Center, Hackensack University Medical Center, Hackensack, NJ, USA; [3]UC San Diego Moores Cancer Center, San Diego, CA, USA; [4]Miami Cancer Institute | Baptist Health South Florida, Miami, FL, USA; [5]Division of Hematology, Oncology and Transplantation, Hôpital Maisonneuve-Rosemont/Université de Montréal, Montréal, Québec, Canada; [6]Pediatric Oncology Branch, National Cancer Institute, Bethesda, MD, USA; [7]Clinical Research Division, Fred Hutchinson Cancer Center, Seattle, WA, USA; [8]Department of Medicine, Division of Oncology, Washington University School of Medicine, St. Louis, MO, USA; [9]Department of Medicine, Adult Bone Marrow Transplant Services, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [10]Hematologics, Seattle, WA, USA; [11]Vor Biopharma, Cambridge, MA, USA.

Objective

To evaluate the safety of trem-cel (formerly known as VOR33) and gemtuzumab ozogamicin (GO; tradename Mylotarg™) in acute myeloid leukemia (AML) patients at a high risk of relapse post-hematopoietic cell transplant (HCT).

Background

- Relapse is the leading cause of death for patients undergoing allogeneic HCT for AML,[1] particularly for patients with high-risk features such as minimal residual disease (MRD).[2,3]
- CD33 is an antigen found on the majority of AML cells, however it is also present on normal hematopoietic cells.[4] Agents targeting CD33, such as GO, an anti-CD33 antibody-drug conjugate, therefore cause extensive cytopenias.[5,6]
- Preclinical animal experiments have previously shown that CD33 is dispensible for normal hematopoiesis.[7]
- In order to reduce AML relapse post-HCT, a *CD33* CRISPR/Cas9 gene-edited donor allograft, trem-cel, was developed to enable post-HCT CD33-directed therapies while protecting healthy donor cells from on-target myelosuppression.

Methods

Figure 1. VBP101 (NCT04849910) Trial Schema

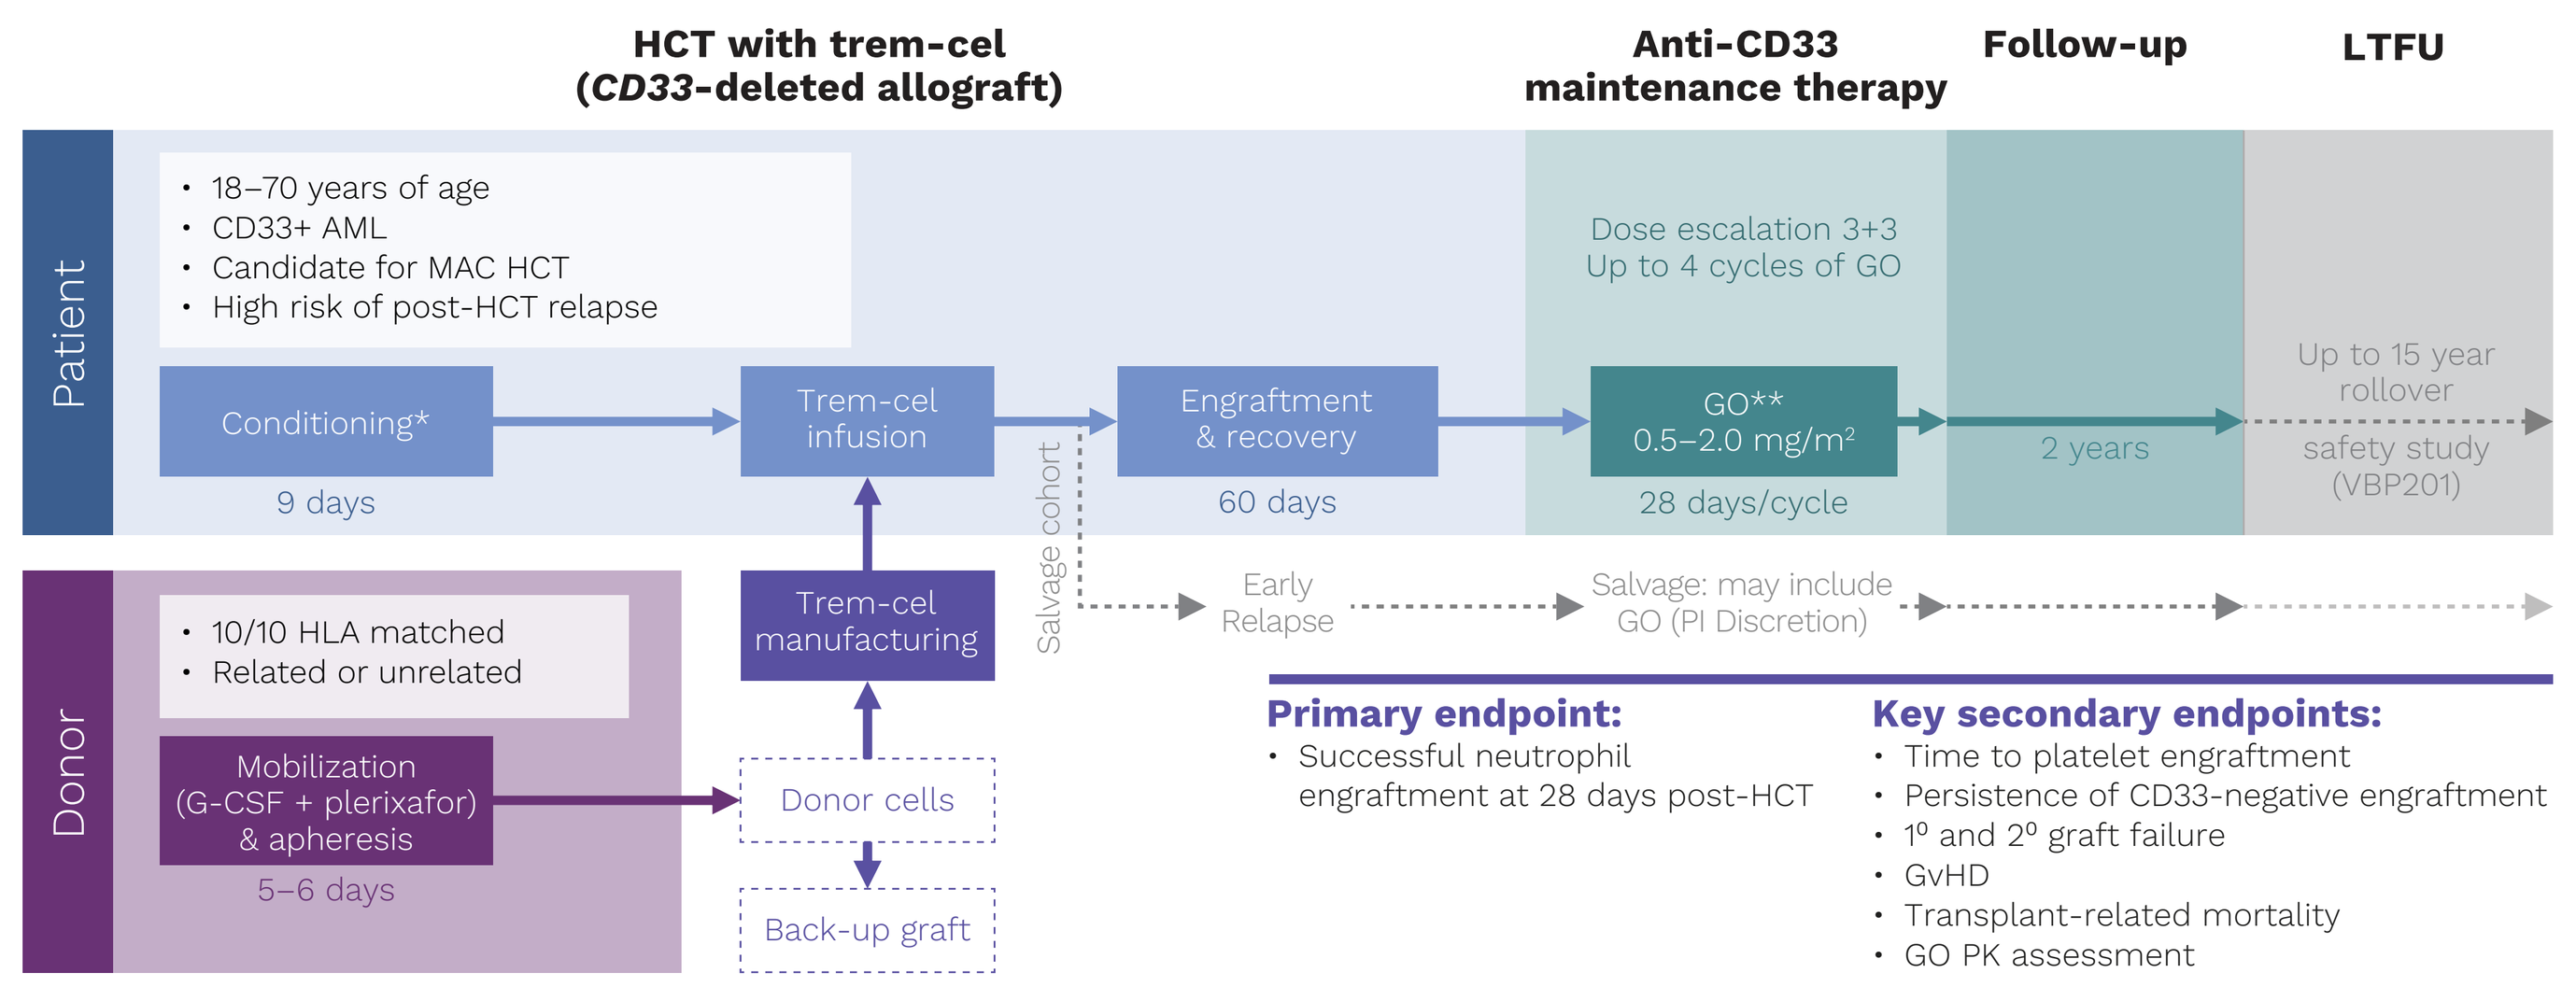
HCT with trem-cel
(CD33-deleted allograft)
Anti-CD33 maintenance therapy
Follow-up
LTFU
Patient
18–70 years of age
CD33+ AML
Candidate for MAC HCT
High risk of post-HCT relapse
Conditioning*
9 days
Trem-cel infusion
Engraftment & recovery
60 days
Dose escalation 3+3
Up to 4 cycles of GO
GO**
0.5–2.0 mg/m²
28 days/cycle
2 years
Up to 15 year rollover
safety study (VBP201)
Salvage cohort
Early Relapse
Salvage: may include GO (PI Discretion)
Donor
10/10 HLA matched
Related or unrelated
Mobilization (G-CSF + plerixafor) & apheresis
5–6 days
Trem-cel manufacturing
Donor cells
Back-up graft
Primary endpoint:
Successful neutrophil engraftment at 28 days post-HCT
Key secondary endpoints:
Time to platelet engraftment
Persistence of CD33-negative engraftment
1º and 2º graft failure
GvHD
Transplant-related mortality
GO PK assessment

*Conditioning consists of busulfan/melphalan/fludarabine/rATG or total body irradiation/cyclophosphamide/thiotepa/rATG; **Dose escalation is executed using a 3+3 design with a total of 3 cohorts. The process of dose escalation is continued until MTD is met and RP2D is determined.

VBP101 is an open-label first-in-human trial, where CD33-positive AML patients who are at high risk of relapse (MRD+ or evidence of persistent BM blasts) undergo myeloablative HCT with trem-cel followed by treatment with low-dose GO **(Figure 1)**. Part 1 of the study will enroll 9–18 evaluable patients in 3 cohorts who will be treated with escalating doses of GO (0.5-2.0 mg/m²) in a 28-day treatment cycle for up to 4 cycles. Part 1 will evaluate the safety of trem-cel and will determine the maximum tolerated dose (MTD) and recommended phase 2 dose (RP2D) of GO. Part 2 will enroll an additional 12 evaluable patients to further evaluate the safety of trem-cel and preliminary efficacy of trem-cel and GO at RP2D.

Results

Patient Characteristics

Patient 1

- 64-year-old female patient diagnosed with AML including high-risk features: highly complex (adverse) cytogenetics, myelodysplasia (MDS)-related changes, and *TP53* mutation.
- Initial treatment consisted of two courses of cytarabine/daunorubicin to achieve complete response (CR). The patient then relapsed and required three cycles of high-dose cytarabine (HiDAC) and two cycles of venetoclax and decitabine to achieve second complete response (CR2), however bone marrow (BM) contained 1.8% MRD.
- A 10/10 HLA matched unrelated donor (MUD) was identified and consented. Donor was mobilized using granulocyte colony stimulating factor (G-CSF) and plerixafor per protocol. Trem-cel was manufactured with 88% *CD33* gene editing efficacy; the patient's trem-cel dose was 7.6 x 10⁶ CD34+ cells/kg.
- The patient received myeloablative conditioning (MAC) with busulfan/melphalan/fludarabine/anti-thymocyte globulin (ATG) prior to trem-cel infusion.

Patient 2

- 32-year-old male diagnosed with AML after myeloid sarcoma partially resected from the small bowel and omentum.
- Initial cytogenetics were Inv 16 and +22. Subsequent t(3;3) was identified (adverse risk).
- Initial treatment consisted of 7+3 cytarabine/daunorubicin with CR but 1.8% MRD in BM. The patient then received 3 cycles of HiDAC, remaining in CR with persistence of abdominal disease by positron emission tomography (PET).
- A 10/10 HLA MUD was identified and consented. Donor was mobilized with G-CSF and plerixafor per protocol. Trem-cel was manufactured with 87% *CD33* gene editing efficacy; the patient's trem-cel dose was 3.2 x 10⁶ CD34+ cells/kg.
- The patient received MAC with busulfan/melphalan/fludarabine/ATG prior to trem-cel infusion.

Data from 6 February 2023 datacut.

Results

Figure 2. Neutrophil Engraftment and Platelet Recovery Post-HCT with Trem-cel

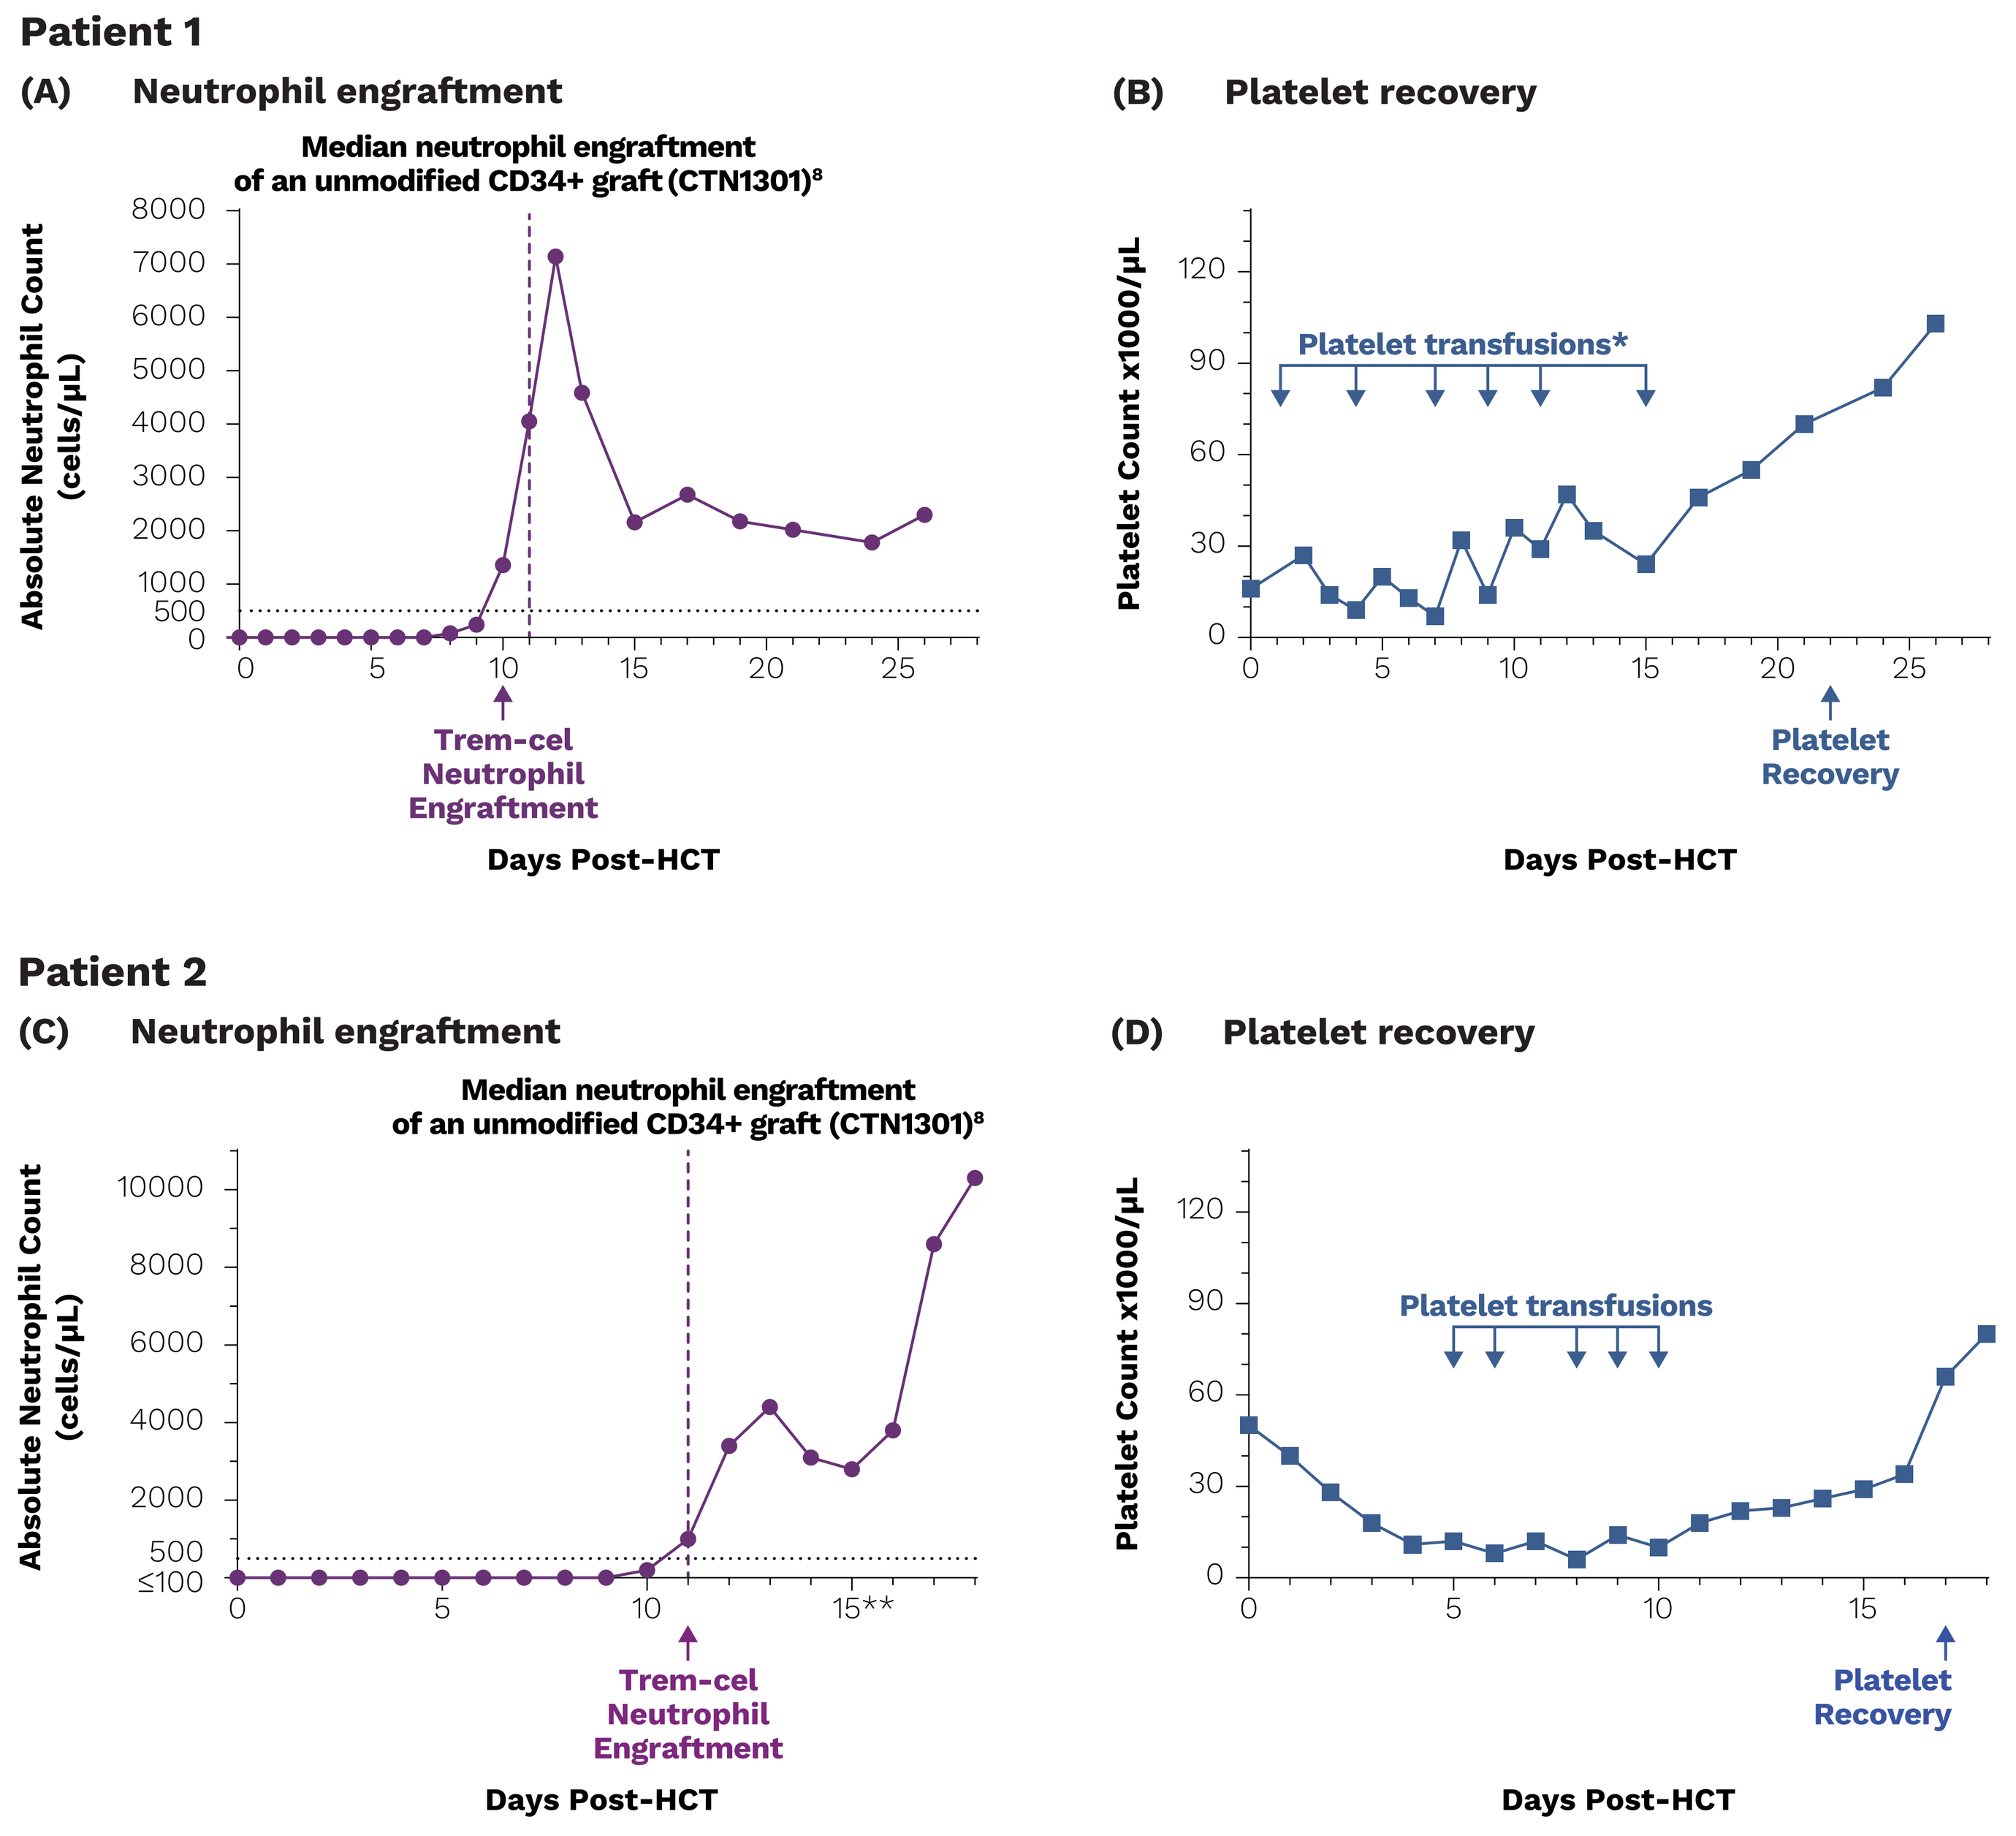
Patient 1
(A) Neutrophil engraftment
Median neutrophil engraftment of an unmodified CD34+ graft (CTN1301)⁸
Absolute Neutrophil Count (cells/μL)
Trem-cel Neutrophil Engraftment
Days Post-HCT
(B) Platelet recovery
Platelet transfusions*
Platelet Count x1000/μL
Platelet Recovery
Days Post-HCT
Patient 2
(C) Neutrophil engraftment
Median neutrophil engraftment of an unmodified CD34+ graft (CTN1301)⁸
Absolute Neutrophil Count (cells/μL)
Trem-cel Neutrophil Engraftment
Days Post-HCT
(D) Platelet recovery
Platelet transfusions
Platelet Count x1000/μL
Platelet Recovery
Days Post-HCT

*Elevated platelet transfusion threshold of 30K/μL used due to pre-existing hemorrhage risk in Patient 1; **Patient 2 received steroids on Days 15–18.

In Patient 1, following trem-cel infusion, neutrophil engraftment and platelet recovery occurred at **10** and **22 days**, respectively (**Figure 2A** and **B**). In Patient 2, neutrophil engraftment occurred at **11 days** while platelet recovery occurred at **17 days** (**Figure 2C** and **D**).

Table 1. Donor Chimerism, CD33 Editing and Expression in Peripheral Blood Post-HCT with Trem-cel (Patient 1)

Transplant Day	D28	D60	D100*
Bulk			
Donor Chimerism	100%	100%	100%
CD33 Gene Editing (Indels)	95.2%	95.9%	99.4%
Monocytes (CD14+ CD15+)			
Donor Chimerism	100%	100%	100%
CD33 Gene Editing (Indels)	95.0%	95.6%	99.7%
% CD33 Negative Cells by Flow	95.3%	96.0%	99.9%
NK cells (CD16+ CD56+)			
Donor Chimerism	100%	100%	100%
CD33 Gene Editing (Indels)	92.1%	94.9%	95.4%
B cells (CD19+)			
Donor Chimerism	–	100%	100%
CD33 Gene Editing (Indels)	–	95.6%	98.5%
T cells (CD3+)			
Donor Chimerism	–	–	97.0%
CD33 Gene Editing (Indels)	–	–	97.6%**

*Two doses of GO were administered between D60 and D100; **97.6% includes all T cells (97% donor T cells and 3% recipient T cells).

Figure 3. Flow Cytometric Analysis of Peripheral Blood and Bone Marrow Post-HCT with Trem-cel (Patient 1)

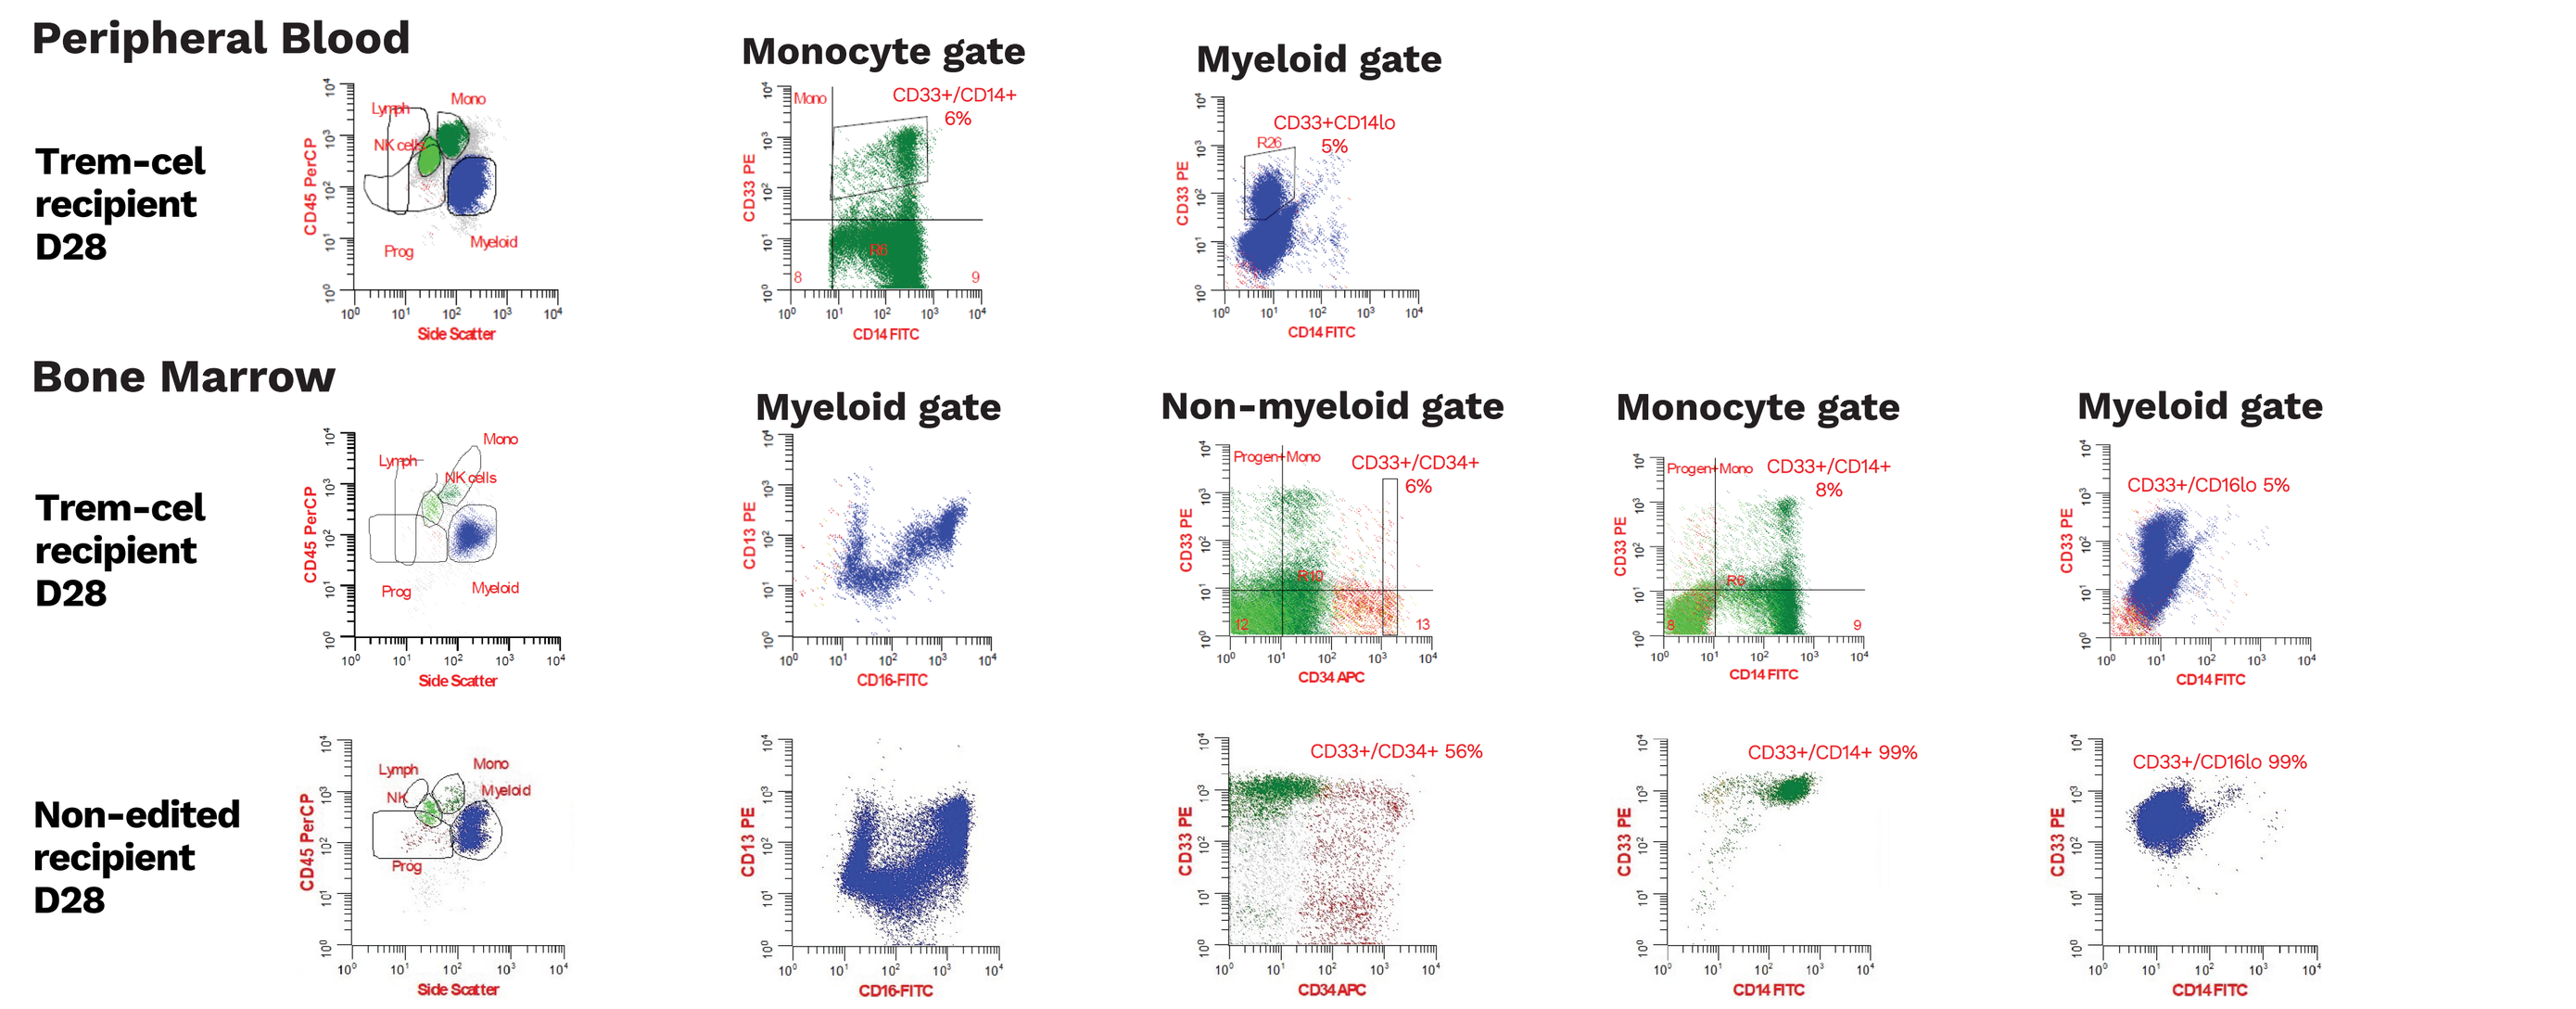
Peripheral Blood
Trem-cel recipient D28
Monocyte gate
Myeloid gate
Bone Marrow
Trem-cel recipient D28
Non-edited recipient D28
Myeloid gate
Non-myeloid gate
Monocyte gate
Myeloid gate
CD33+/CD14+ 6%
CD33+CD14lo 5%
CD33+/CD34+ 6%
CD33+/CD14+ 8%
CD33+/CD16lo 5%
CD33+/CD34+ 56%
CD33+/CD14+ 99%
CD33+/CD16lo 99%

At the Day (D) 28 assessment, flow cytometry of peripheral blood (PB) demonstrated 94% of monocytes and 95% of neutrophils were CD33-negative. BM analysis showed 95% of maturing myeloid cells, 92% of maturing monocytes, and 94% of CD34+ myeloblasts were CD33-negative with development patterns comparable to a reference non-edited graft after an ATG-containing conditioning regimen and 28 days post-HCT (Data provided by M. Loken; **Figure 3**).

Table 2. Pharmacokinetics of GO (0.5mg/m², single infusion) in Presence of CD33-Edited Graft Compared to GO Doses in Relapsed/Refractory AML patients with CD33 (Patient 1)

	VBP101	Relapsed/Refractory AML population (GO phase 1 study 0903A1-101-US)[9]							
Parameter	**0.5 mg/m²**	**0.25 mg/m²**	**0.5 mg/m²**	**1 mg/m²**	**2 mg/m²**	**4 mg/m²**	**5 mg/m²**	**6 mg/m²**	**9 mg/m²**
$T_{1/2}$ (Hr)	64.56	–	–	–	–	–	–	–	–
T_{max} (Hr)	2.95	–	–	–	–	–	–	–	–
C_{max} (ng/mL)	259	15	28	50	411	611	1,325	2,219	2,870
AUC_{last} (Hr*ng/mL)	19049.14	–	–	–	–	–	–	–	–
AUC_{inf} (Hr*ng/mL)	22923.70	82	468	943	11,110	10,970	29,980	69,300	80,430
Vz (L/m**2)	2.03	–	–	–	–	–	–	–	–
CL (L/hr/m**2)	0.0218	–	–	–	–	–	–	–	–

The patient received GO (0.5 mg/m²) at 68 days post-HCT during the anti-CD33 maintenance therapy period of the trial. Plasma samples were collected pre-GO dosing and post-GO dosing on Cycle (C)1D1 (1, 2, 3, 4, and 6 hours) and C1D8 and analyzed for hP67.6 by enzyme-linked immunosorbent assay (ELISA). Pharmacokinetic (PK) parameters were calculated using a non-compartmental analysis method in Phoeniz WinNonlin Version 9.3 using the concentration data measured after drug infusion and the actual dosing and PK sampling information **(Table 2)**.

Table 3. Absolute Neutrophil and Platelet Counts as well as CD33-Negative Monocytes and Neutrophils Pre- and Post-GO Treatment (Patient 1)

	Post-HCT Recovery		Cycle 1 GO (0.5 mg/m²)						Cycle 2 GO (0.5 mg/m²)				Cycle 3 GO (0.5 mg/m²)			
Transplant Day	**D28**	**D60**	**D68**	**D73**	**D76**	**D84**	**D87**	**D94**	**D96**	**D101**	**D108**	**D116**	**D124**	**D129**	**D136**	**D147**
GO Cycle Visit Day			**C1D1***	**C1D6**	**C1D9**	**C1D17**	**C1D20**	**C1D27**	**C2D1***	**C2D5**	**C2D12**	**C2D20**	**C3D1***	**C3D6**	**C3D13**	**C3D24**
Absolute Neutrophil Count (Cells/μL)	2,300	2,770	2,260	3,140	3,490	3,330	3,250	3,400	3,570	3,640	4,510	2,450	3,170	2,900	2,290	2,150
Platelet Count (x 1000 Cells/μL)	103	120	91	118	120	110	94	95	99	100	96	97	95	94	76	74
CD33-Negative Neutrophils (%)	95	96	96	–	–	–	–	–	>99.9	>99.9	–	–	>99.9	–	–	99
CD33-Negative Monocytes (%)**	94	94	93	–	–	–	–	–	97	>99.6	–	–	98.8	–	–	94.9

*The C1D1, C2D1, and C3D1 PB assessments were conducted prior to GO infusion; **Monocyte flow overlapping with CD33+ leukemic blast population.

Figure 4. Neutrophil and Platelet Counts After GO Doses (0.5 mg/m²) (Patient 1)

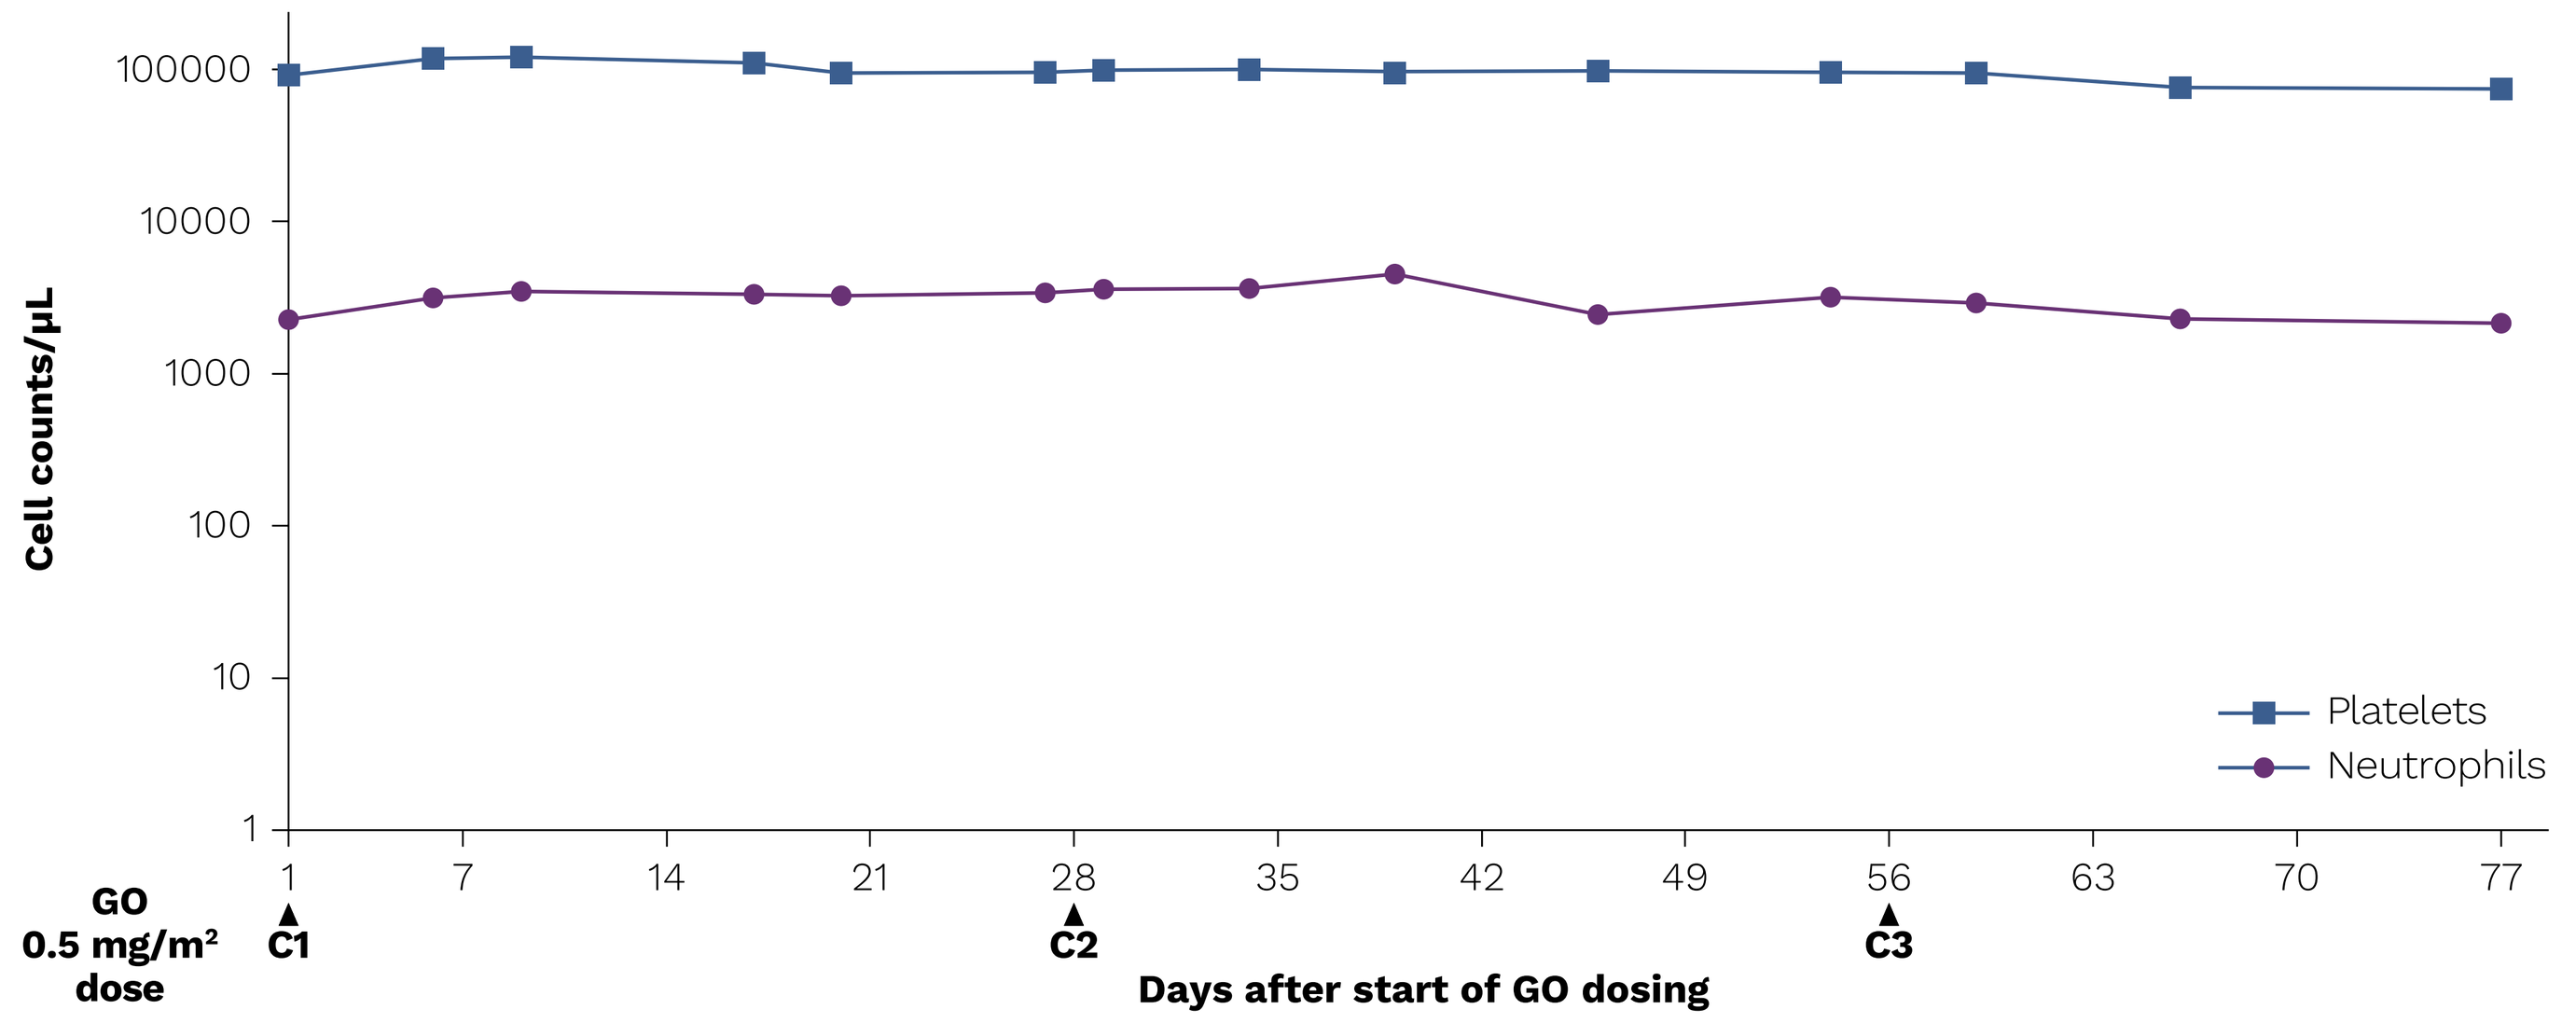
Cell counts/μL
Platelets
Neutrophils
GO 0.5 mg/m² dose
C1
C2
C3
Days after start of GO dosing

Following dosing with GO (0.5 mg/m²) at D68 post-HCT, neutrophil and platelet counts were stable in the patient through D141 **(Figure 4)**. No elevations in liver function tests (LFTs) were observed.

Clinical Course and Safety

Patient 1

- The patient was discharged from the transplant unit on D15. She received GO (0.5 mg/m²) every 28 days beginning at D68 post-HCT for a total of 3 doses. Neutrophil and platelet counts during GO doses are reported in **Table 3** and **Figure 4**.
- Serious adverse events (SAEs) after trem-cel dosing included Grade 3 renal colic attributable to nephrolithiasis.
- Infectious adverse events (AEs) included a Grade 1 and Grade 2 skin infection, Grade 2 cytomegalovirus (CMV) reactivation, Grade 2 urinary tract infection, and Grade 2 BK virus (urine); all resolved or resolving.
- Hepatic AEs included Grade 1 and Grade 2 aspartate aminotransferase (AST)/alanine aminotransferase (ALT) elevations; both were attributed to anti-fungal therapy and resolved.
- No trem-cel-related AEs were reported. GO-related AEs included Grade 1 nausea and Grade 2 vomiting.
- Grade 2 acute graft-versus-host disease (aGvHD) of the gut was reported and is responding to non-systemic steroids.
- Prior to HCT, the patient had 1.8% MRD in the BM and none was observed post-HCT at D28 and D60. GO was started at D68. MRD in the BM was detected at D101 (0.3%) and D129 (2.2%). The patient discontinued study treatment after Cycle 3 of GO. D147 BM showed relapsed disease with 7.6% blasts.

Patient 2

- Infectious AEs post-trem-cel dose included Grade 3 febrile neutropenia and *Escherichia coli (E. coli)* bacteremia reported at D8 prior to engraftment.
- No SAEs were reported after trem-cel dose through D18.
- No trem-cel-related AEs were reported.

Conclusions

- **In this first-in-human trial, results from the first two patients transplanted with trem-cel demonstrated neutrophil engraftment and platelet recovery similar to patients who received non-edited CD34 selected grafts.[8]**
- **Trem-cel was well-tolerated, with no related AEs and no unexpected AEs reported.**
- **A high level of CD33-negative hematopoiesis was achieved (>90%) and increased over the course of GO treatment. The pattern of myeloid development derived from the *CD33*-deleted graft was similar to a reference patient with a non-edited graft.**
- **CD33-negative cells were enriched after GO treatment across lineages. The *CD33* gene deletion was observed at the genetic level across all lineages, suggesting editing of an early progenitor hematopoietic cell.**
- **In the context of a *CD33*-deleted graft, a 0.5 mg/m² dose of GO demonstrated a maximum GO concentration (C_{max}) and area under the curve (AUC) in plasma comparable to doses of 1–2 mg/m² and 4–5 mg/m², respectively, in AML patients.**
- **At a GO dose of 0.5 mg/m², neutrophil and platelet counts remained stable through multiple cycles of GO, including in the presence of MRD. This suggests protection from GO hematotoxicity.**
- **These initial data support the biological dispensability of CD33 in myeloid development and a potential approach enabling post-HCT treatment with GO and other CD33-targeted therapies.**

AE: adverse event; **aGvHD:** acute graft-versus-host disease; **ALT:** alanine aminotransferase; **AML:** acute myeloid leukemia; **AST:** aspartate aminotransferase; **ATG:** anti-thymocyte globulin; **AUC_{inf}:** area under the curve from time = 0 extrapolated to infinity; **AUC_{last}:** area under the curve from time = 0 to the last measurable time point; **BM:** bone marrow; **C:** cycle; **CL:** clearance; **C_{max}:** maximum concentration; **CMV:** cytomegalovirus; **CR:** complete response; **CR2:** second complete response; **D:** day; ***E. coli:*** *Escherichia coli*; **ELISA:** enzyme-linked immunosorbent assay; **G-CSF:** granulocyte-colony stimulating factor; **GO:** gemtuzumab ozogamicin; **GvHD:** graft-versus-host disease; **HCT:** hematopoietic cell transplant; **HiDAC:** high-dose cytarabine; **HLA:** human leukocyte antigen; **LFT:** liver function test; **LTFU:** long-term follow-up; **MAC:** myeloablative conditioning; **MDS:** myelodysplasia; **MRD:** minimal residual disease; **MTD:** maximum tolerated dose; **MUD:** matched unrelated donor; **NK:** natural killer; **PB:** peripheral blood; **PET:** positron emission tomography; **PI:** principal investigator; **PK:** pharmacokinetic; **rATG:** rabbit anti-thymocute globulin; **RP2D:** recommended phase 2 dose; **SAE:** serious adverse event; **$T_{1/2}$:** half-life; **T_{max}:** time taken to reach the maximal concentration or time to C_{max}; **Vz:** volume of distribution.

References: [1]Tsirigotis P. et al. Bone Marrow Transplant 2016;51(11):1431–1438; [2]Norkin M. et al. Blood Cancer J 2017;7(12):634; [3]Hourigan CS. et al. J Clin Oncol 2020;38(12):1273–1283; [4]Kenderian SS. et al. Leukemia 2015;29(8):1637–1647; [5]Stein EM. et al. Blood 2018;131(4):387–396; [6]Walter RB. et al. Expert Opin Investig Drugs 2018;27(4):339–348; [7]Borot F. et al. PNAS 2019;116(24):11978–11987; [8]Luznik L. et al. J Clin Oncol 2022;40(4):356–368 [9]Mylotarg ODAC 2017. **Acknowledgements:** This study was funded by Vor Biopharma. We thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. The authors acknowledge Costello Medical for medical writing, and the Design Team at Costello Medical for graphic design assistance. All costs associated with development of this poster were funded by Vor Biopharma. **Disclosures:** Author disclosures are available on the conference website.

Presented at 2023 Tandem Meetings | Transplantation & Cellular Therapy Meetings of ASTCT and CIBMTR, 15–19 February 2023