Gene-Edited Hematopoietic Stem Cells to Enable Next-Generation CAR-T Cell Therapy for the Treatment of AML

Julia Etchin, Yonina Keschner, Mariana Silva, Hillary Hoyt, Matthew Ung, Amanda Halfond, Julia DiFazio, Nate Manalo, Juliana Xavier-Ferrucio, Michelle Lin, Gary Ge, Julian Scherer, John Lydeard, Tirtha Chakraborty

Vor Bio, 100 Cambridgepark Dr, Cambridge, MA, USA 02140

#1775

INTRODUCTION

- Targeted immunotherapy of Acute Myeloid Leukemia (AML) has been limited due to lack of tumor-specific antigens resulting in "on-target, off-tumor" effects that can lead to severe cytopenia
- To unlock the full potential of targeted treatments, we engineer hematopoietic stem cells (HSCs) by genetically ablating target antigens from healthy, donor-derived HSCs for hematopoietic stem cell transplant. This allows compatible immuno-therapy to specifically kill leukemic cells bearing the AML target-antigen while sparing the antigen-null allogenic graft[1-4]
- Combinatorial targeting of AML antigens may be required to avoid antigen escape and address tissue heterogeneity, while multiplex genome editing of these antigens is necessary to protect the transplanted healthy graft (Figure 1)

OBJECTIVE

- Profile prospective AML antigens in a longitudinal assessment of AML patient bone marrow samples
- Investigate ADGRE2 antigen as a prospective AML target and evaluate novel ADGRE2-targeting CAR-Ts
- Assess ADGRE2 expression in healthy heme tissues to evaluate risk of on-target, off-tumor toxicity
- Evaluate ADGRE2 biological dispensability and demonstrate its multiplexing with other targets in healthy HSCs to enable combinatorial targeting of AML cells while sparing normal cells

Figure 1. Vor Multiplex Platform

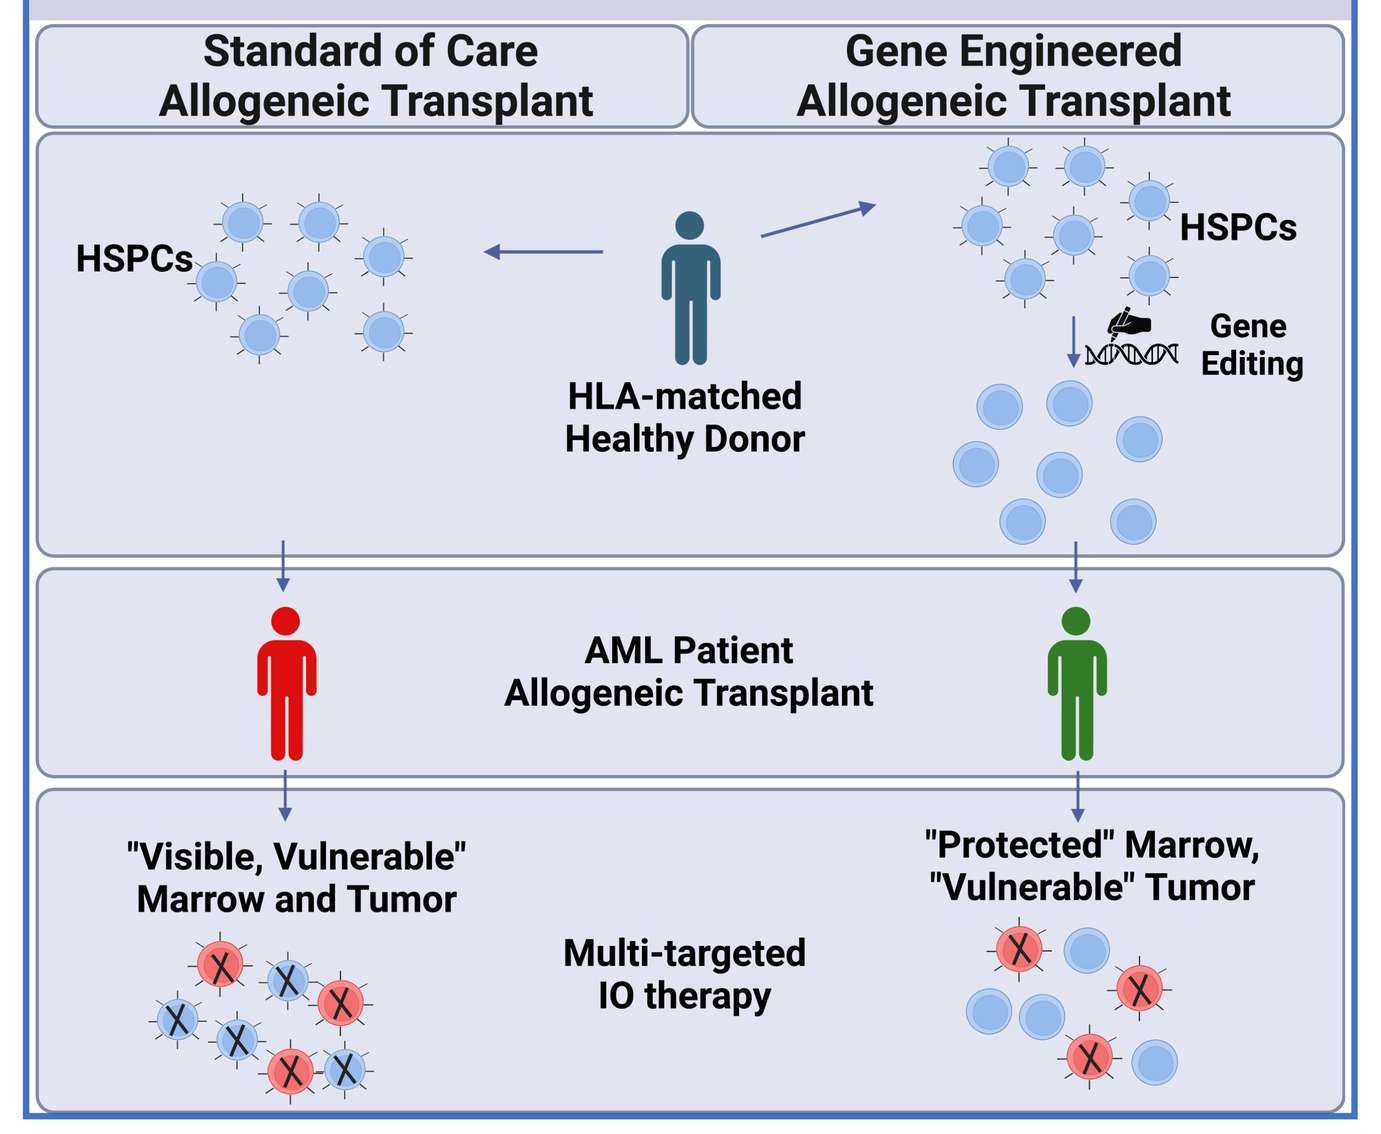
Standard of Care Allogeneic Transplant
Gene Engineered Allogeneic Transplant
HSPCs
HLA-matched Healthy Donor
HSPCs
Gene Editing
AML Patient Allogeneic Transplant
"Visible, Vulnerable" Marrow and Tumor
Multi-targeted IO therapy
"Protected" Marrow, "Vulnerable" Tumor

Figure 2. ADGRE2/EMR2 Structure and Background

- Member of the epidermal growth factor (EGF) – seven-transmembrane (TM7) sub-family (ADGRE1, 2, 3, 4, and 5 or CD97) of Adhesion G protein-coupled receptor (GPCR) family[5]
- Alternative splicing results in multiple isoforms that contain varying number of EGF domains
- Plays a role in adhesion, migration, invasion, angiogenesis, and inflammatory response[6-8]
- Expressed on healthy myeloid cells and the majority of AML blasts and leukemic stem cells (LSCs)[9]
- High occurrence of loss-of-function (LOF) alleles reported in the gnomAD database[10]
- A gain-of-function mutation within the GAIN domain of ADGRE2 causes familial vibratory urticaria, a rare autosomal dominant dermal allergic disorder resulting from vibration-induced mast cell activation[11-12]

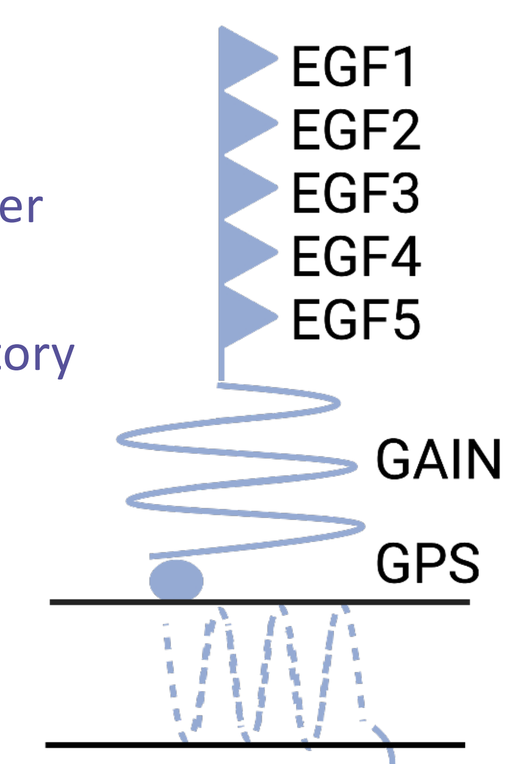
EGF1
EGF2
EGF3
EGF4
EGF5
GAIN
GPS

Results: Novel Immunotherapeutic Targeting of ADGRE2

Figure 3. ADGRE2 is Expressed on AML Blasts and Leukemia Stem Cells

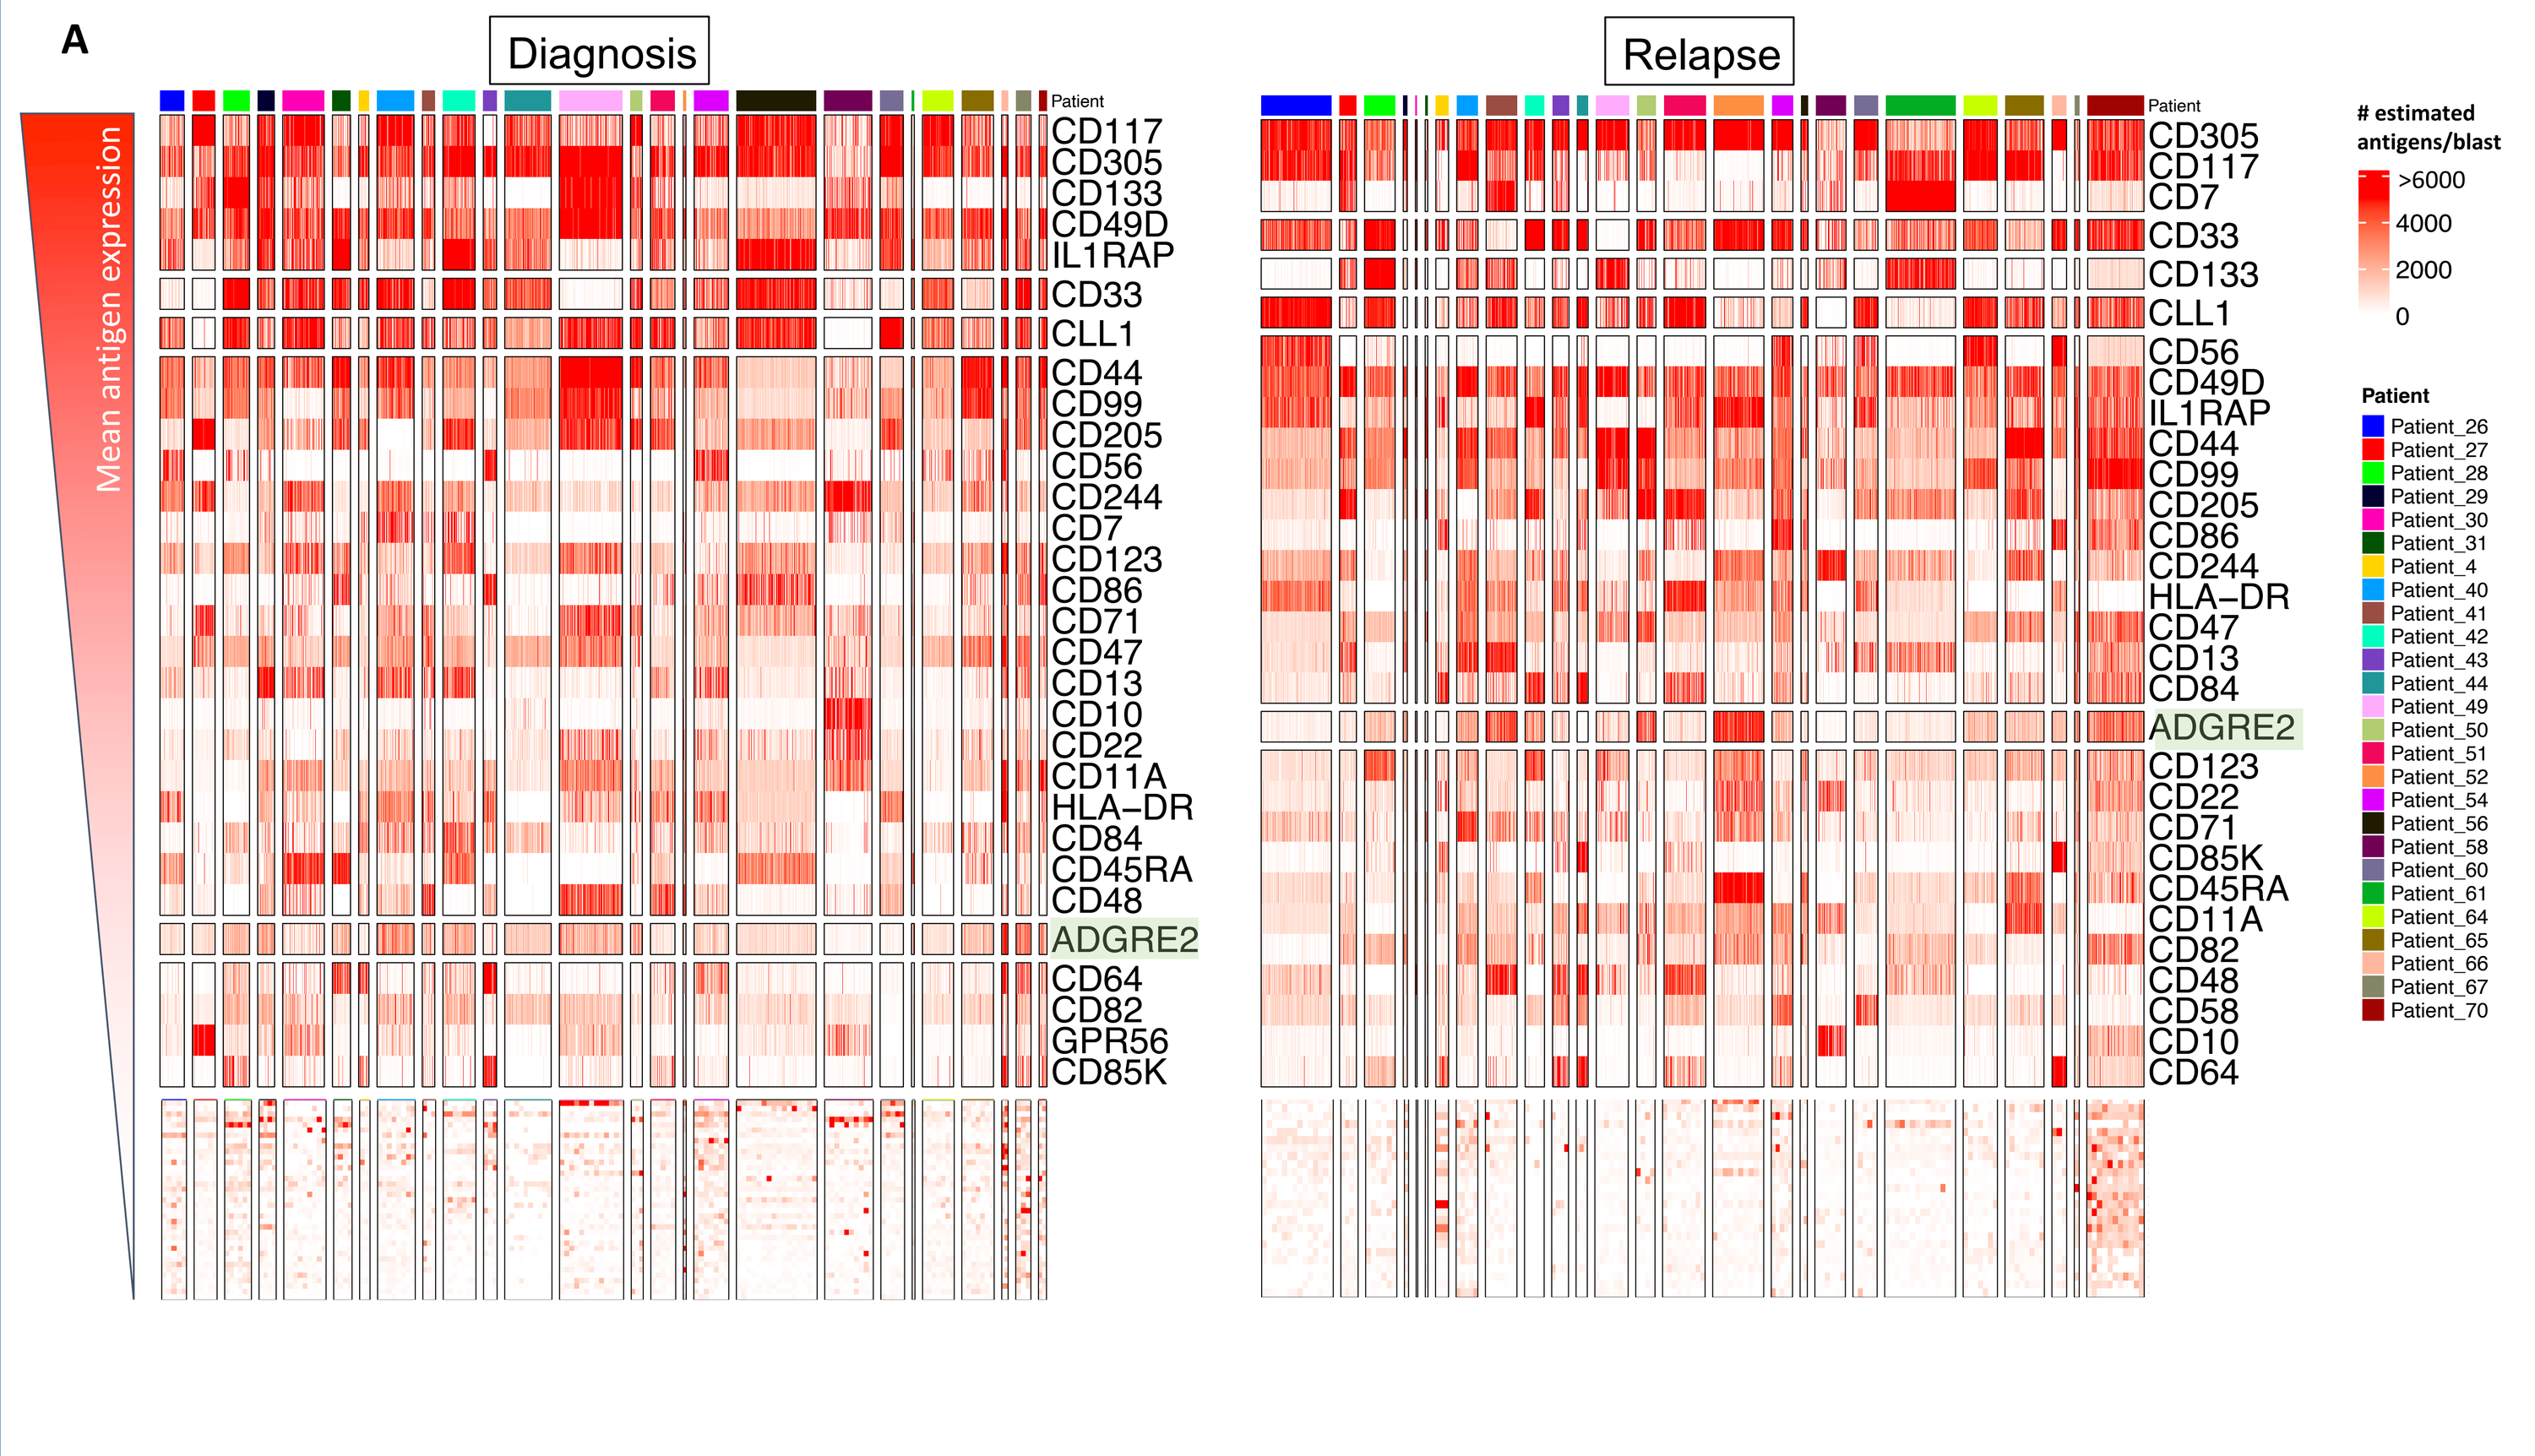
A
Diagnosis
Relapse
Mean antigen expression
estimated antigens/blast
>6000
4000
2000
0
Patient
ADGRE2

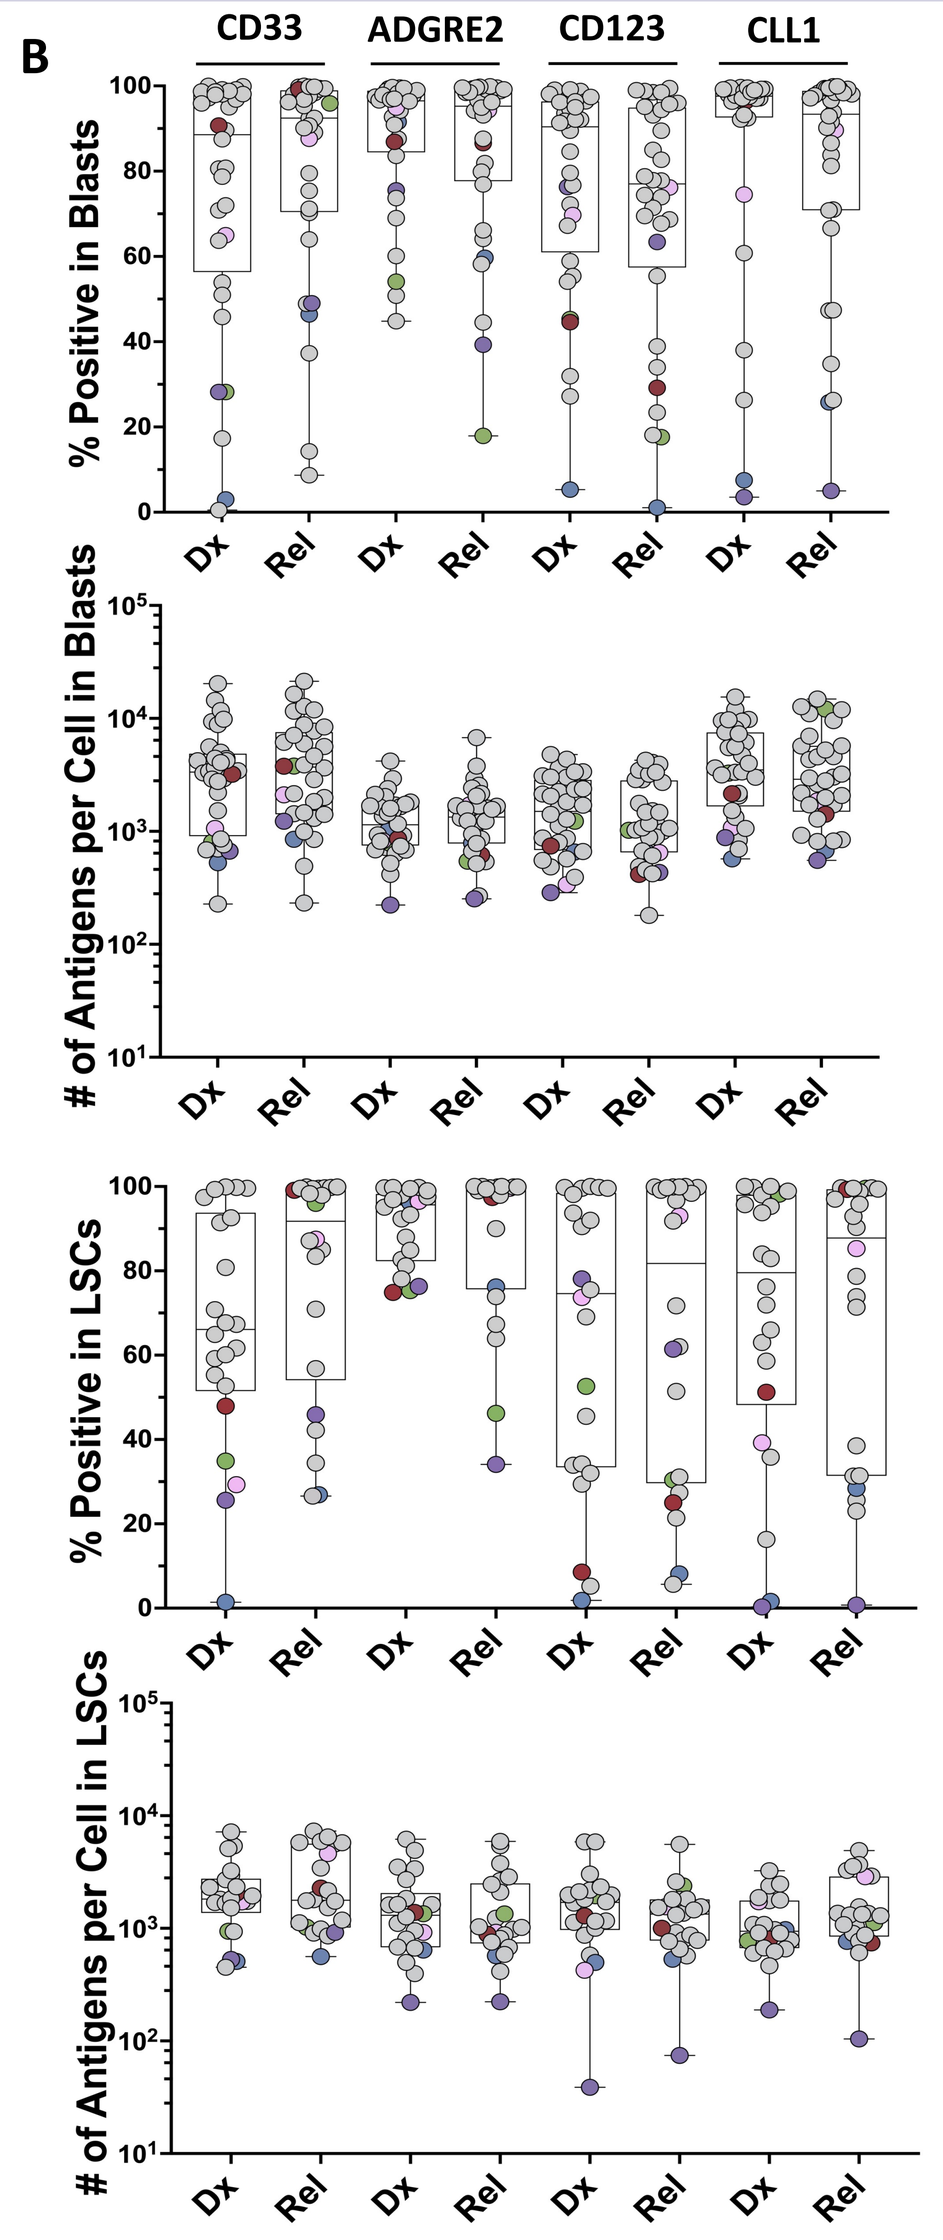
B
CD33
ADGRE2
CD123
CLL1
% Positive in Blasts
of Antigens per Cell in Blasts
% Positive in LSCs
of Antigens per Cell in LSCs
Dx
Rel

A. Cellular indexing of transcriptomes and epitopes (CITE-seq) profiling of 81 candidate antigens across 400,000 cells from 28 paired diagnosis and relapse patient bone marrow samples (BMs). A promising AML target candidate, ADGRE2/EMR2, is among the top 30 highest surface expressors in both diagnosis and relapsed samples. This multimodal profiling was paired with flow cytometry surface expression analysis. **B**. Percent positivity and number of antigens per antigen⁺ cell in blasts and LSCs. Quantibrite PE beads (BD Biosciences) were used for antigen quantitation. Colored dots trace expression data for select patients to demonstrate expression pattern for the four antigens. Data shown as median ± standard error of mean. Leukemic stem cells (LSCs), Diagnosis (Dx), Relapse (Rel).

Figure 4. ADGRE2 -directed CAR-Ts mediate potent specific cytolytic activity against AML cells *in vitro*

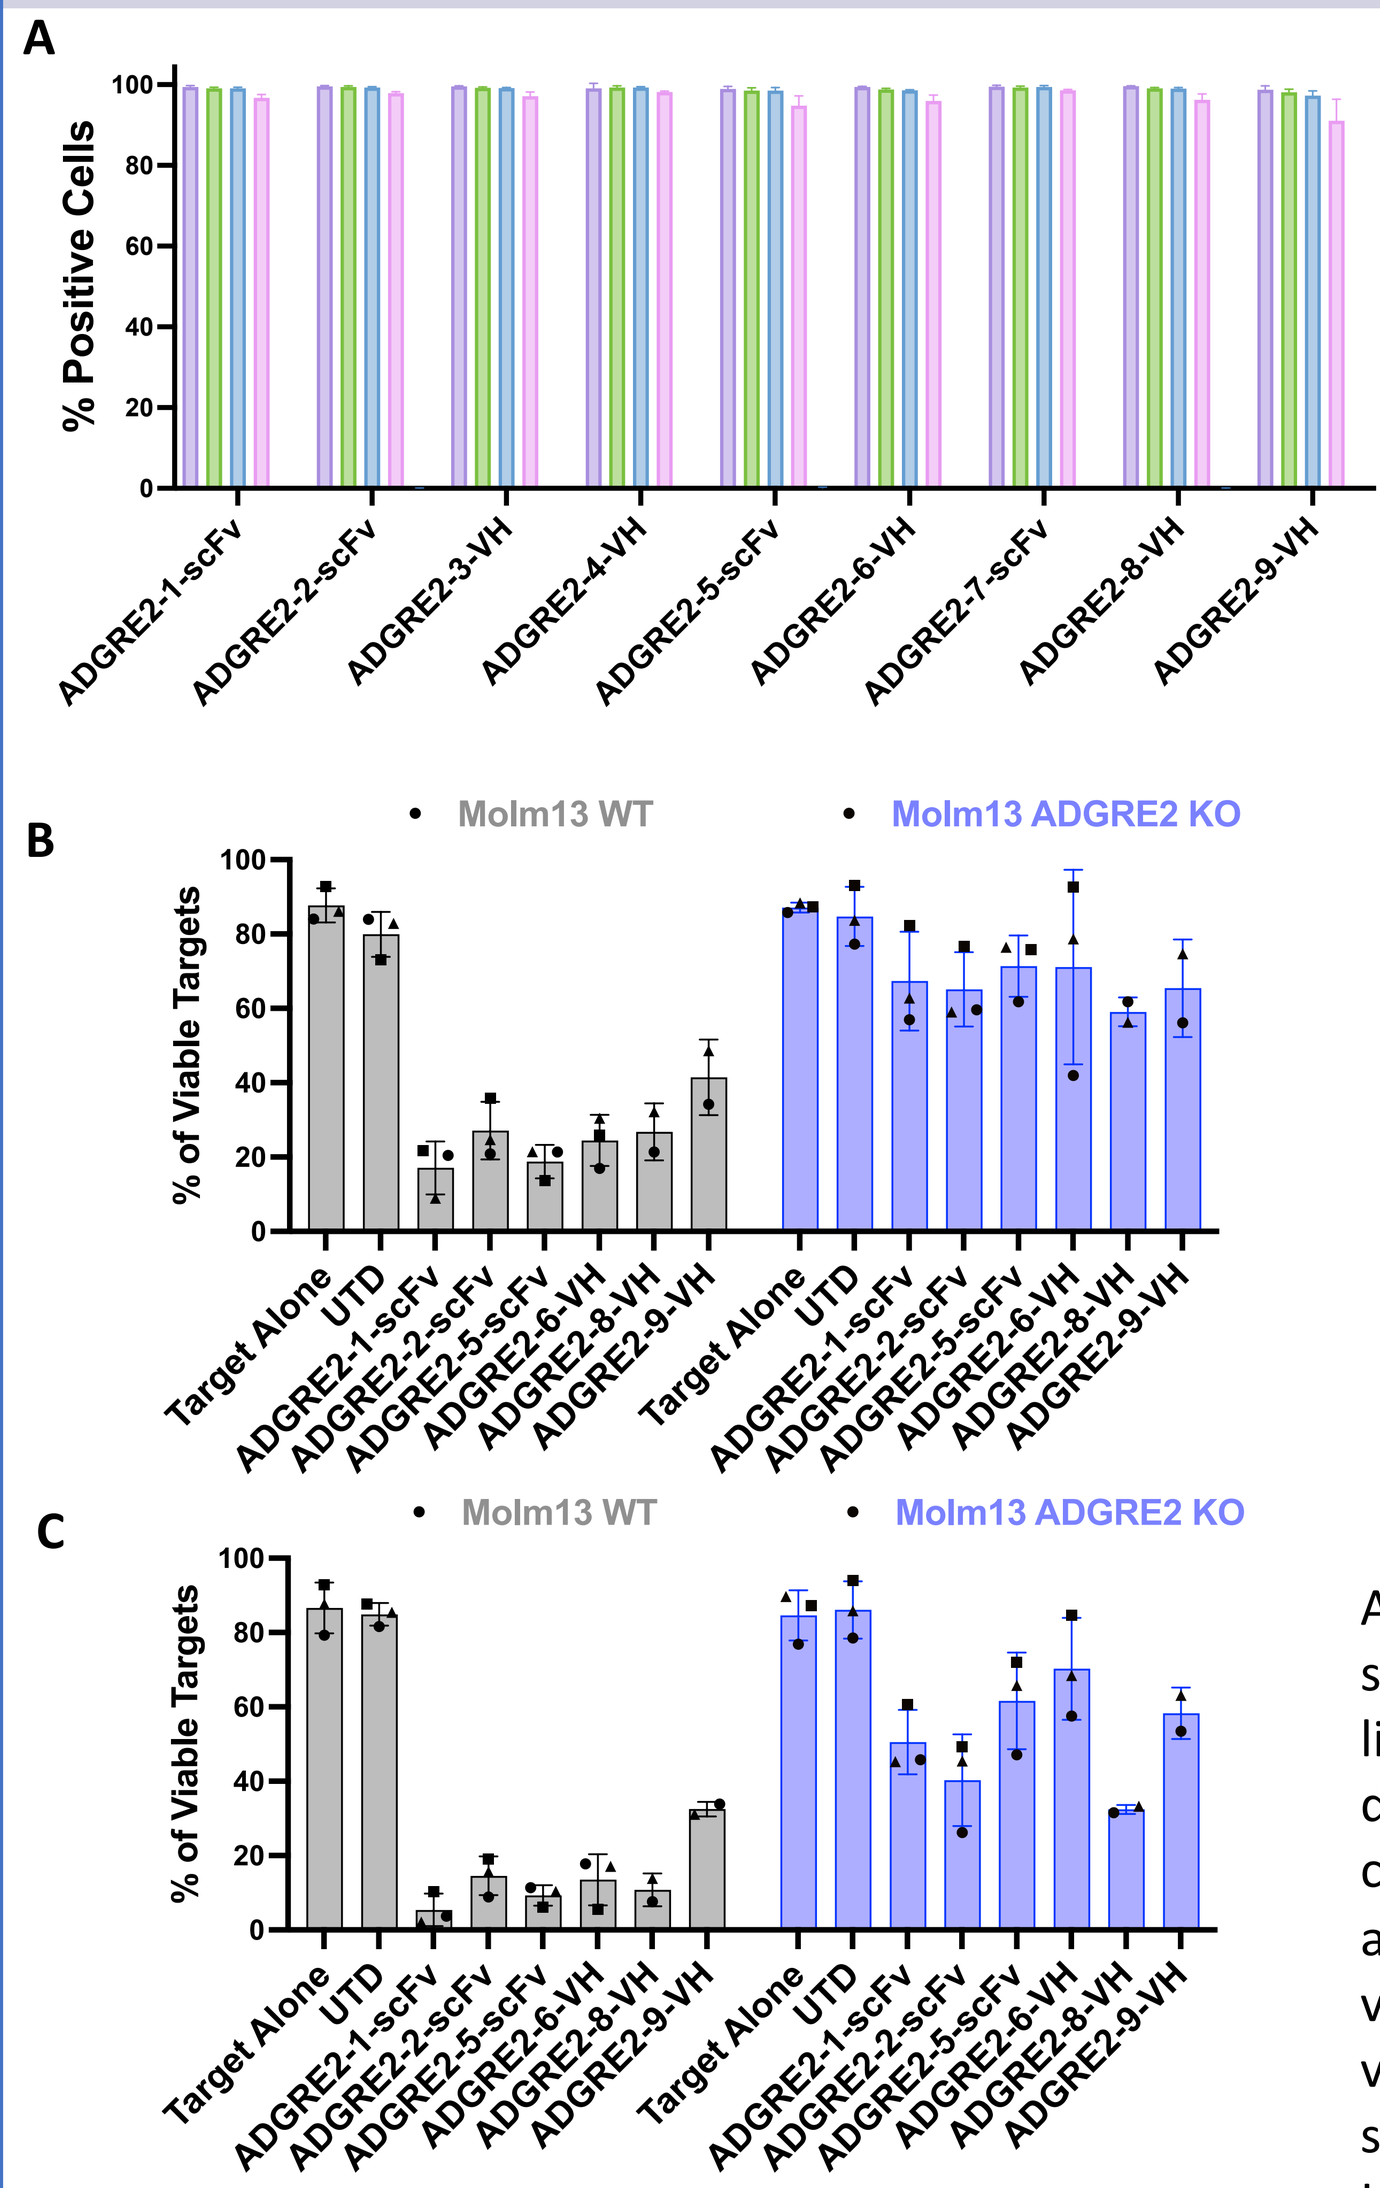
A
% Positive Cells
ADGRE2FL
ADGRE2ΔEGF4
ADGRE2ΔEGF3-4
ADGRE2ΔEGF3-5
CD97FL
B
C
Molm13 WT
Molm13 ADGRE2 KO
% of Viable Targets
Target Alone
UTD

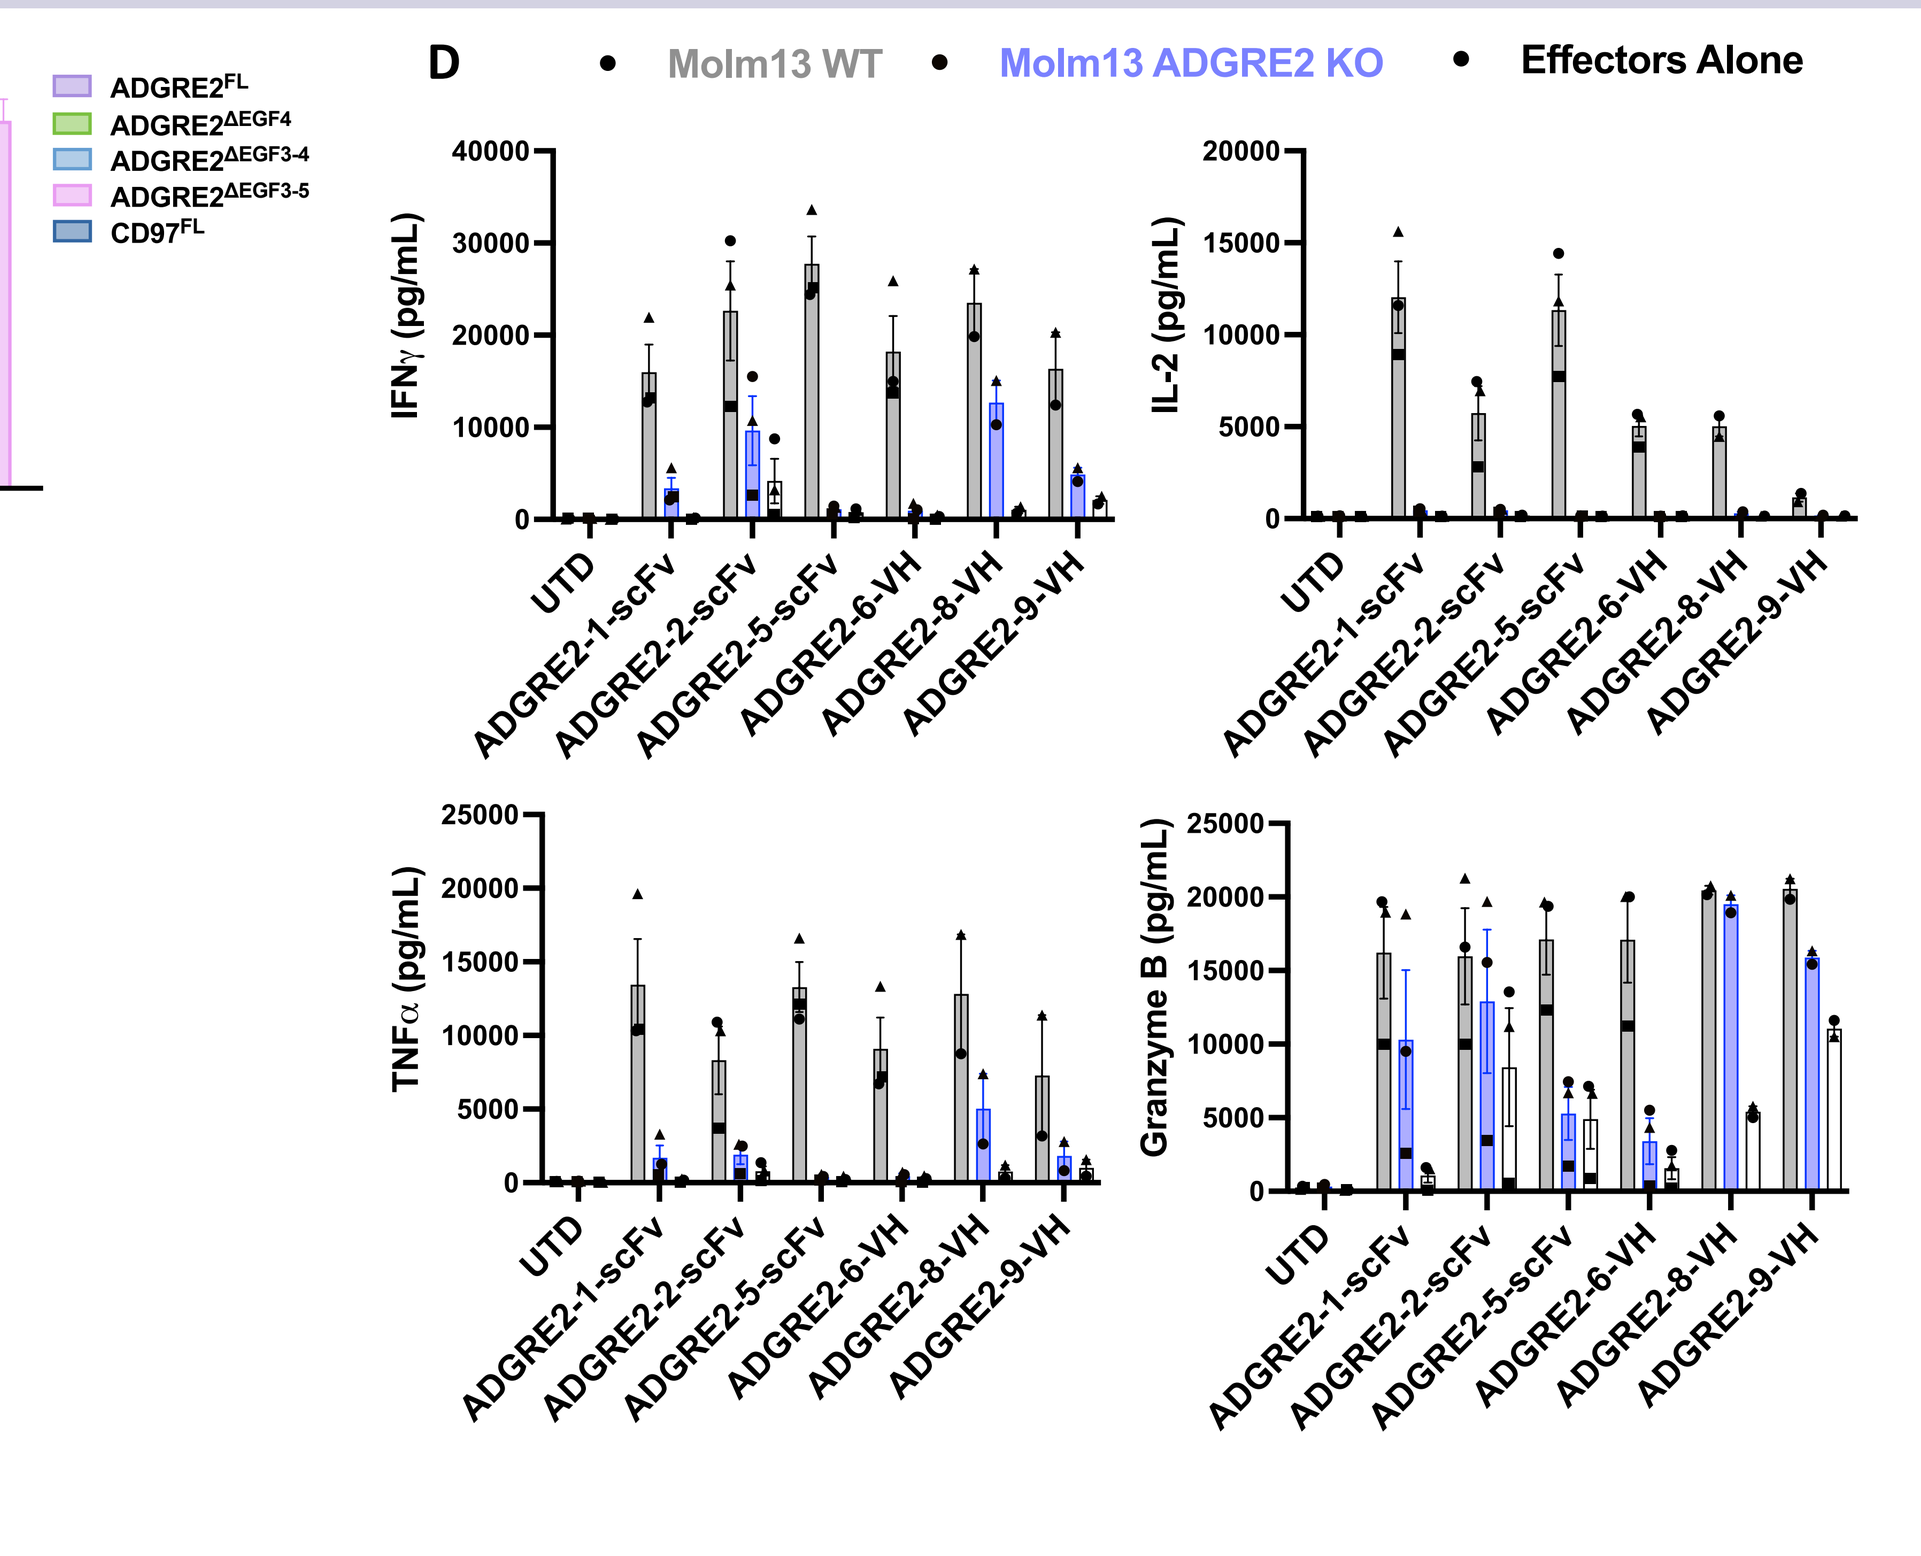
D
Molm13 WT
Molm13 ADGRE2 KO
Effectors Alone
IFNγ (pg/mL)
IL-2 (pg/mL)
TNFα (pg/mL)
Granzyme B (pg/mL)

ADGRE2-directed binders were identified by phage display panning against single-chain variable fragment (scFv) and heavy chain variable region (V_H) libraries. **A.** Flow cytometric analysis of ADGRE2-directed binder reactivity to different ADGRE2 isoforms and CD97 in 293T cells. **B-D.** ADGRE2-directed CAR T cells were co-cultured with CFSE labeled MOLM13 WT and ADGRE2-KO cell lines as AML target cells at 1:1 ratio for 24 and 48 hours (n=3). **B-C.** Percentage of viable target cells was determined by the absence of Annexin V and fixable viability dye reactivity by flow cytometry at 24 **(B)** and 48 **(C)** hours. **D.** The supernatants of co-cultures were analyzed for cytokine secretion using a Luminex 17-Plex kit. Data shown as median ± standard error of mean.

Genome Engineering and Characterization of Edited HSPCs

Figure 5. ADGRE2 is Expressed on Healthy Hematopoietic Stem and Progenitors (HSPCs) and Myeloid Cells

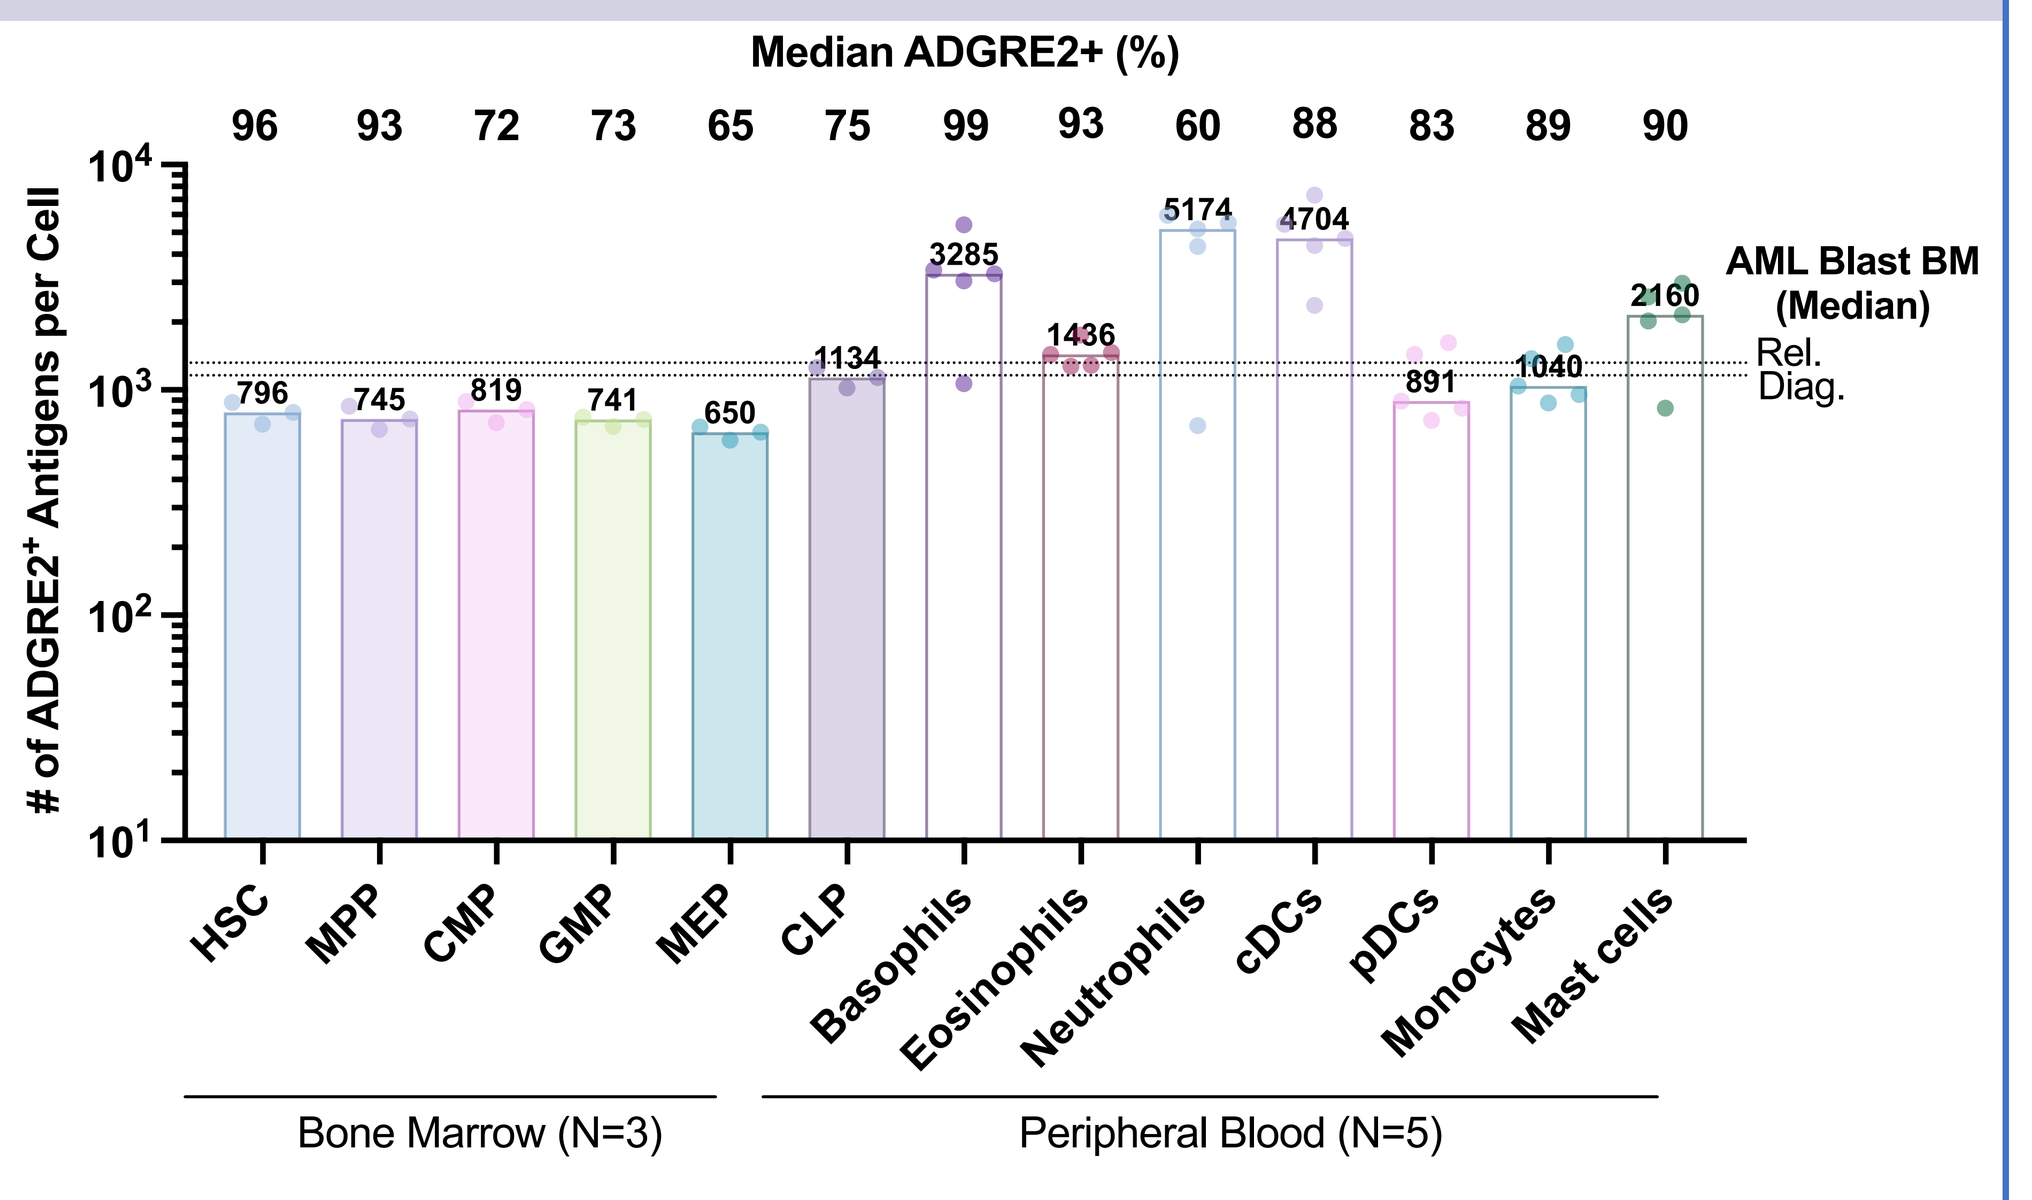
Median ADGRE2+ (%)
96 93 72 73 65 75 99 93 60 88 83 89 90
of ADGRE2+ Antigens per Cell
AML Blast BM (Median)
Rel.
Diag.
HSC MPP CMP GMP MEP CLP Basophils Eosinophils Neutrophils cDCs pDCs Monocytes Mast cells
Bone Marrow (N=3)
Peripheral Blood (N=5)

Flow cytometric analysis of ADGRE2 expression on healthy donor bone marrow and peripheral blood. QuantiBrite beads were used for antigen quantitation. Data shown as mean ± standard deviation. Hematopoietic stem cell (HSC), multipotent progenitor (MPP), common myeloid progenitor (CMP), granulocyte-monocyte progenitor (GMP), megakaryocytic-erythroid progenitor (MEP), common lymphoid progenitor (CLP), basophils, eosinophils, neutrophils, cDCs, pDCs, monocytes, and mast cells.

Figure 6. Experimental Validation of Naturally Occurring ADGRE2 Genetic Variants

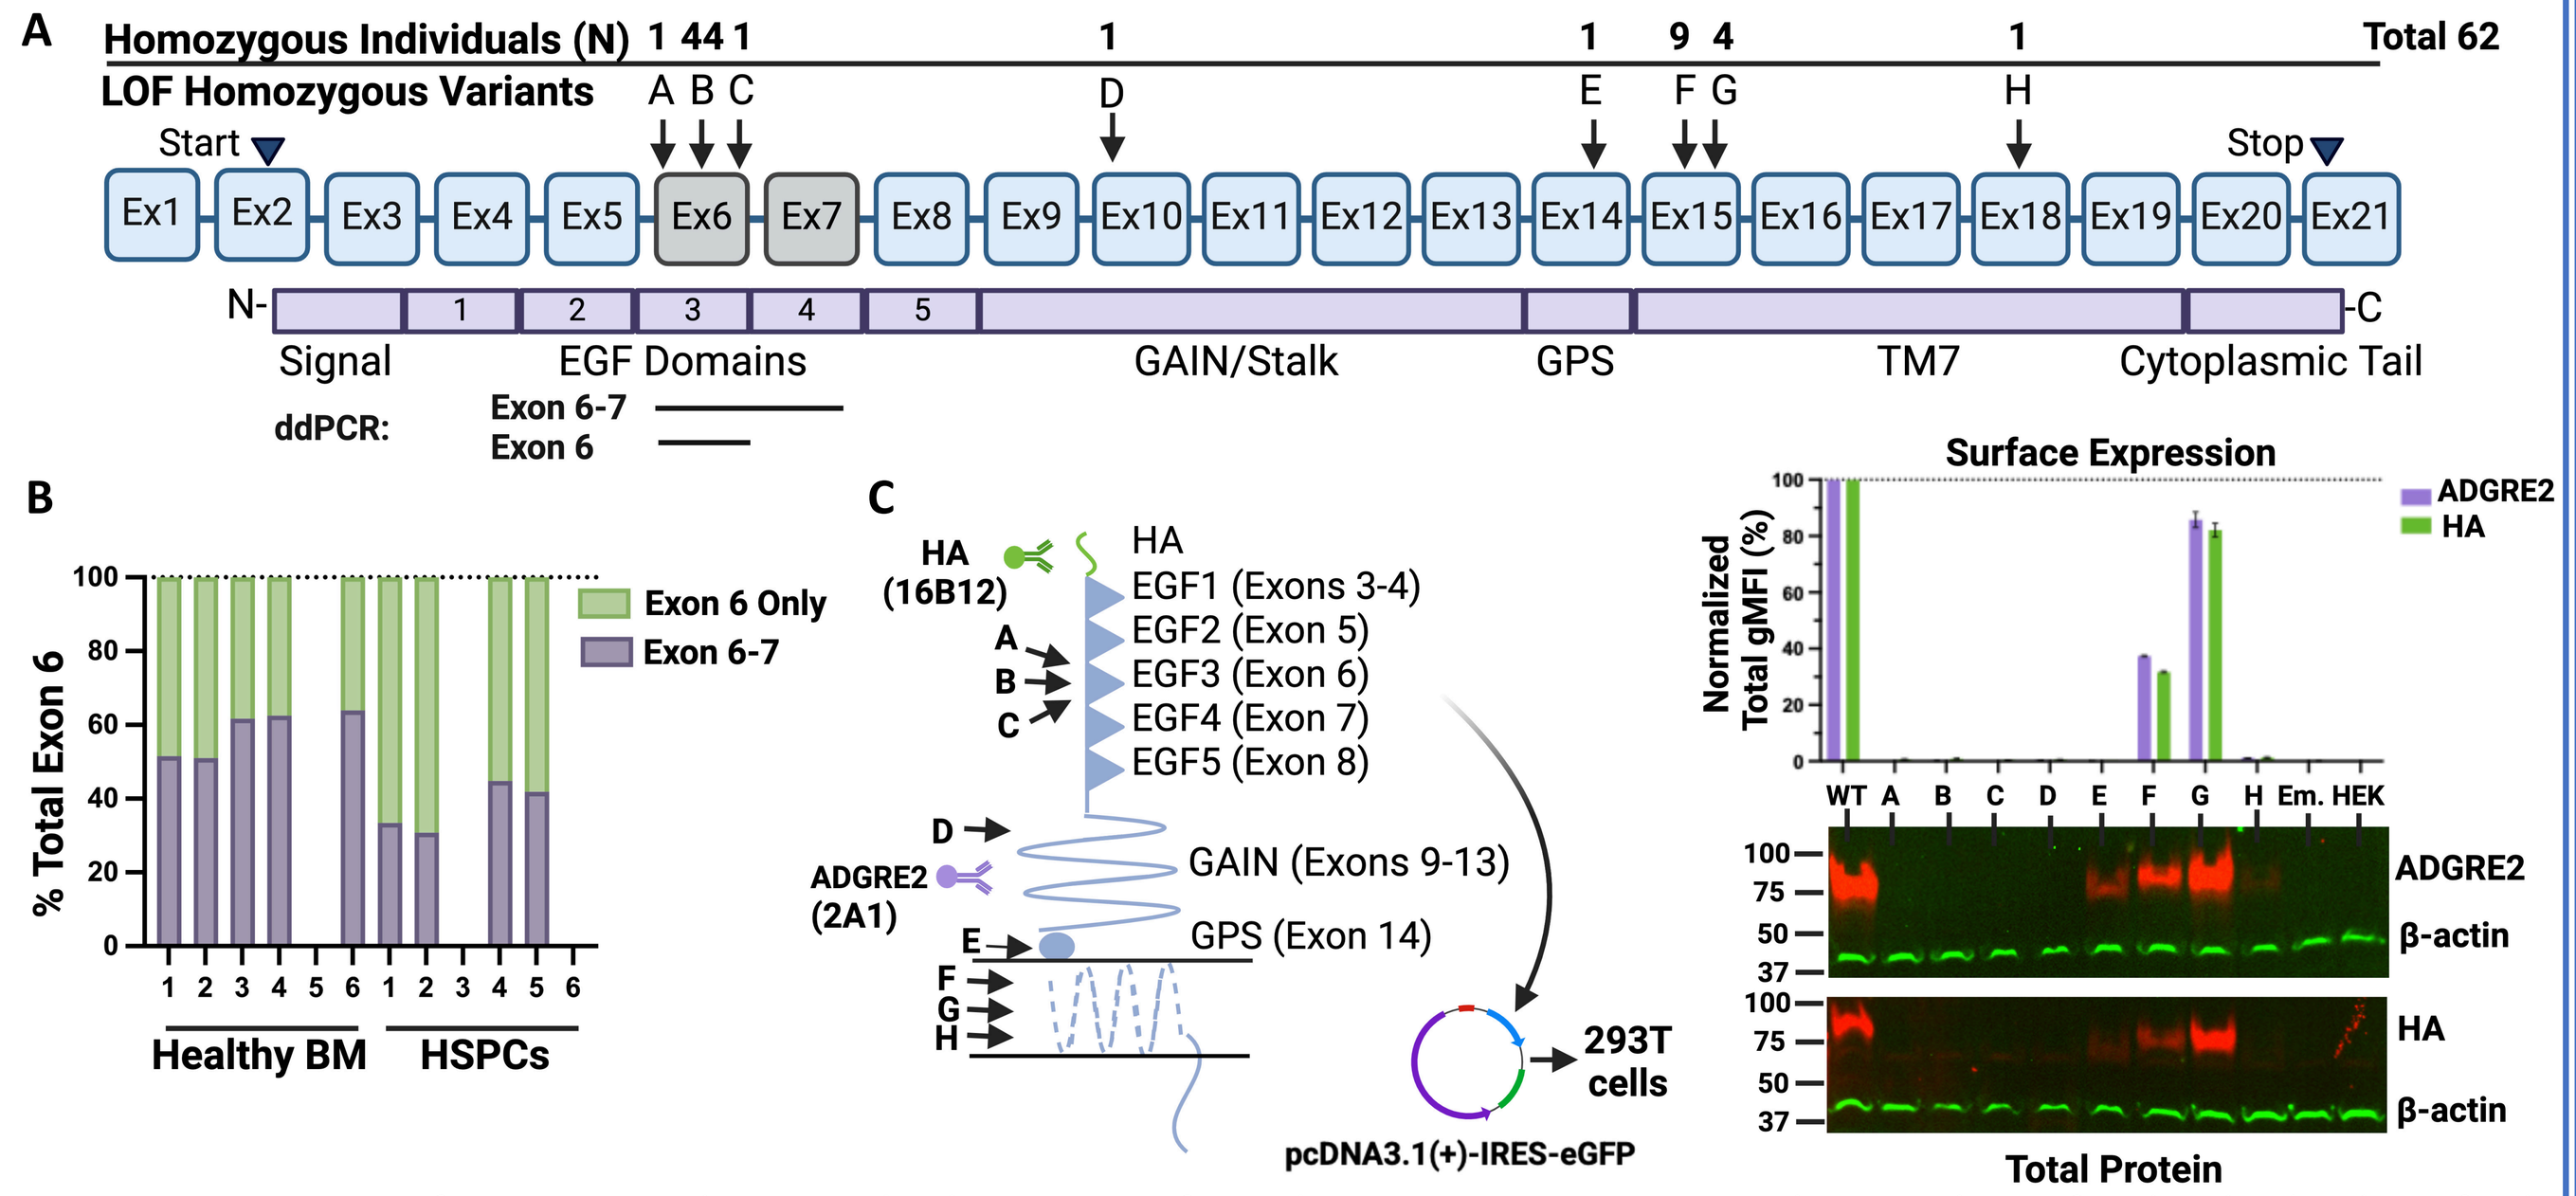
A
Homozygous Individuals (N)
LOF Homozygous Variants
Total 62
Start
Stop
Signal
EGF Domains
GAIN/Stalk
GPS
TM7
Cytoplasmic Tail
ddPCR:
Exon 6-7
Exon 6
B
% Total Exon 6
Exon 6 Only
Exon 6-7
Healthy BM
HSPCs
C
HA (16B12)
HA
EGF1 (Exons 3-4)
EGF2 (Exon 5)
EGF3 (Exon 6)
EGF4 (Exon 7)
EGF5 (Exon 8)
GAIN (Exons 9-13)
GPS (Exon 14)
ADGRE2 (2A1)
293T cells
pcDNA3.1(+)-IRES-eGFP
Surface Expression
Normalized Total gMFI (%)
Total Protein
β-actin

A. Gene structure of *ADGRE2* indicating the locations of LOF homozygous variants. The number of individuals identified for homozygous LOF variants[10] (A, B, C, D, E, F, G, and H) is shown. **B**. Presence and distribution of exon 6 only and exon 6-7 containing isoforms in healthy bone marrow (BM) and hematopoietic stem and progenitor cells (HSPCs) as analyzed by ddPCR (amplified regions indicated in **A**.) **C.** Surface and total protein expression of ADGRE2 variants. Plasmid constructs with ADGRE2 variant mutations were transfected into 293T cells. ADGRE2 and HA antibodies were used for surface and total protein detection by flow cytometry and Western blotting, respectively.

Figure 7. *ADGRE2*-edited HSPCs Engraft Long-term, Differentiate into Multilineages, and Persist *In Vivo*

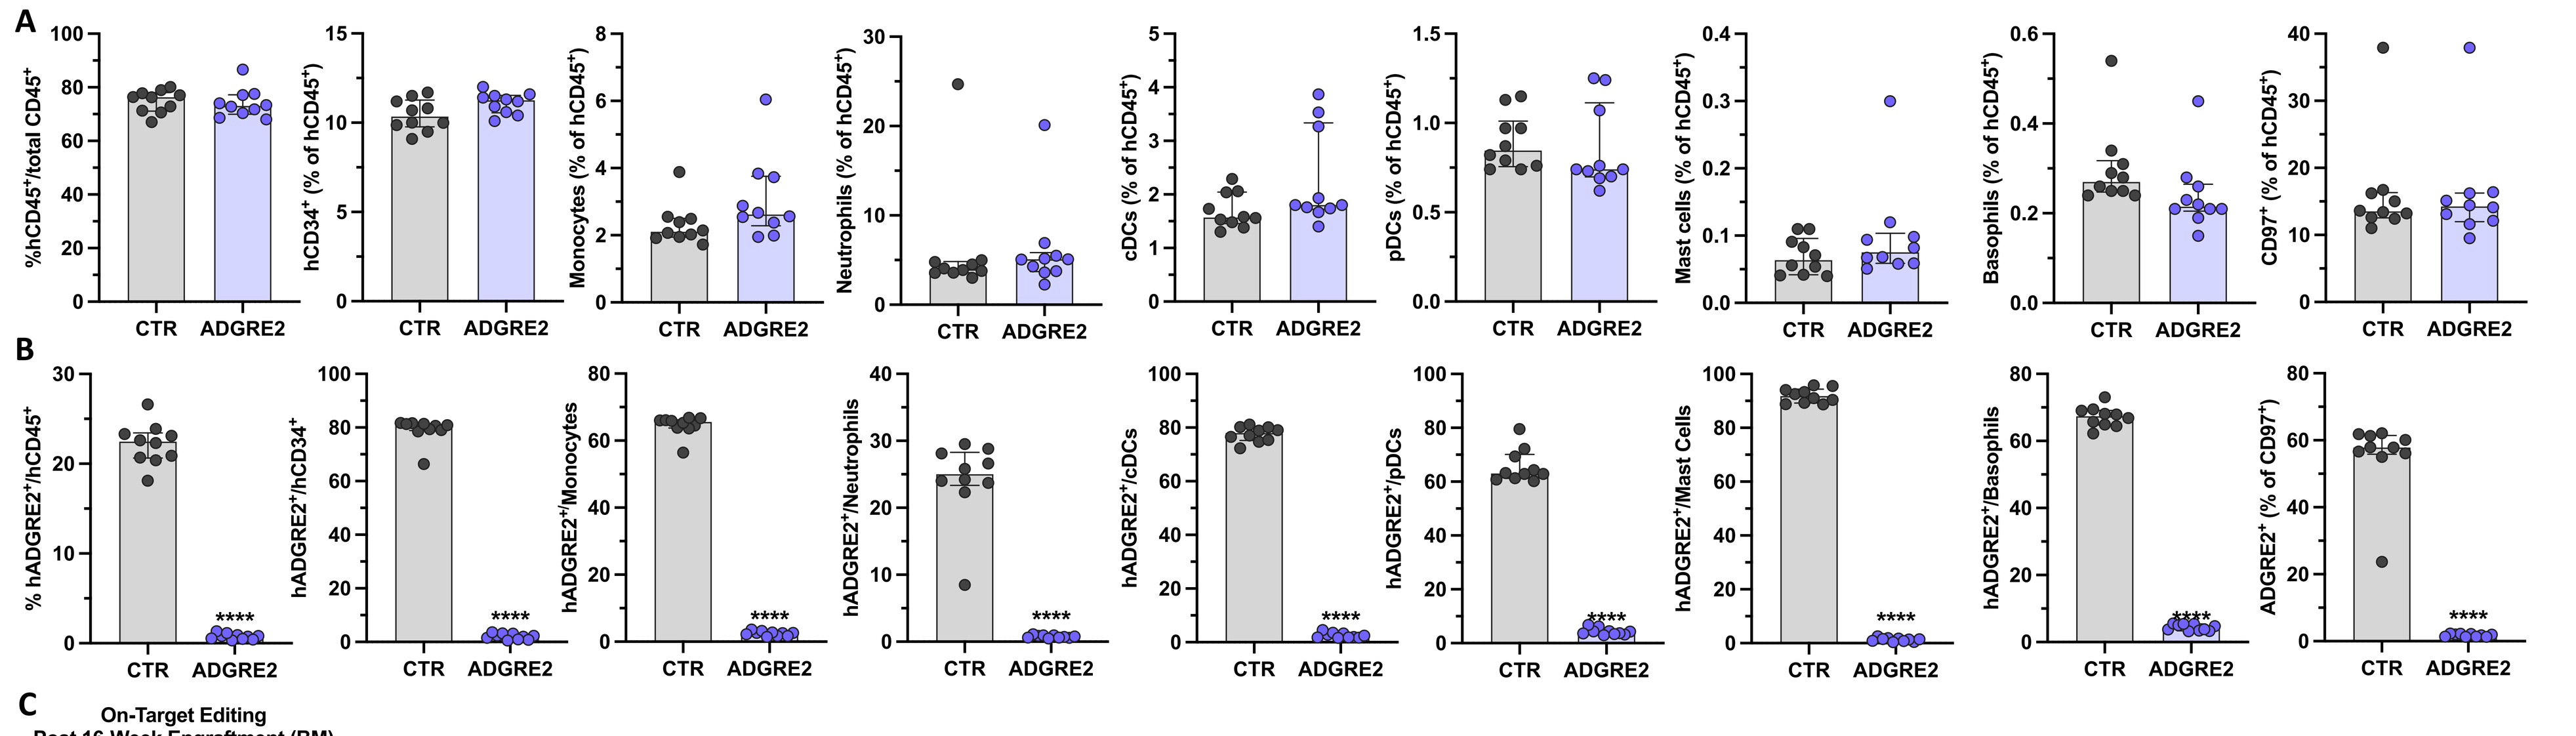
A
B
CTR
ADGRE2

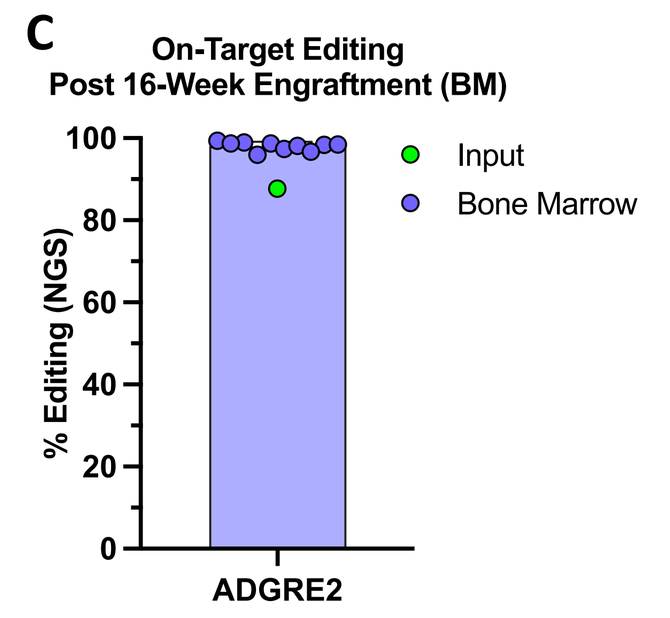
C
On-Target Editing Post 16-Week Engraftment (BM)
% Editing (NGS)
Input
Bone Marrow
ADGRE2

A-C. Control (CTR) unedited or ABE guide *ADGRE2*-edited CD34+ hHSPCs were xenotransplanted into NOD-scid IL2Rg null (NSG) mice followed by 16-week post-engraftment evaluation of the bone marrow (BM). **A.** Flow cytometry analysis of BM showed comparable levels of total human leukocyte chimerism, human HSPCs and various human hematopoietic lineages, between mice engrafted with CTR or edited cells. This indicates that deletion of *ADGRE2* did not impact HSC function, defined by the ability for long-term engraftment and multilineage reconstitution. **B.** Significant reduction of the ADGRE2 protein expression was observed. **C.** Quantification of on-target editing by Next-Gen Sequencing (NGS) of PCR amplicons shows no loss of total editing frequencies after 16 weeks of engraftment at *ADGRE2* locus. The editing frequency of BM samples from each individual mouse are plotted along with the input samples used to engraft the mice. N=10 mice per group. Data shown as mean ± standard deviation.**** p<0.0001 compared to CTR by Unpaired t test.

Figure 8. Efficient Multiplex Adenosine Base Editing of *ADGRE2 in* HSPCs to Support the "Protected" Marrow, "Vulnerable" Tumor Concept

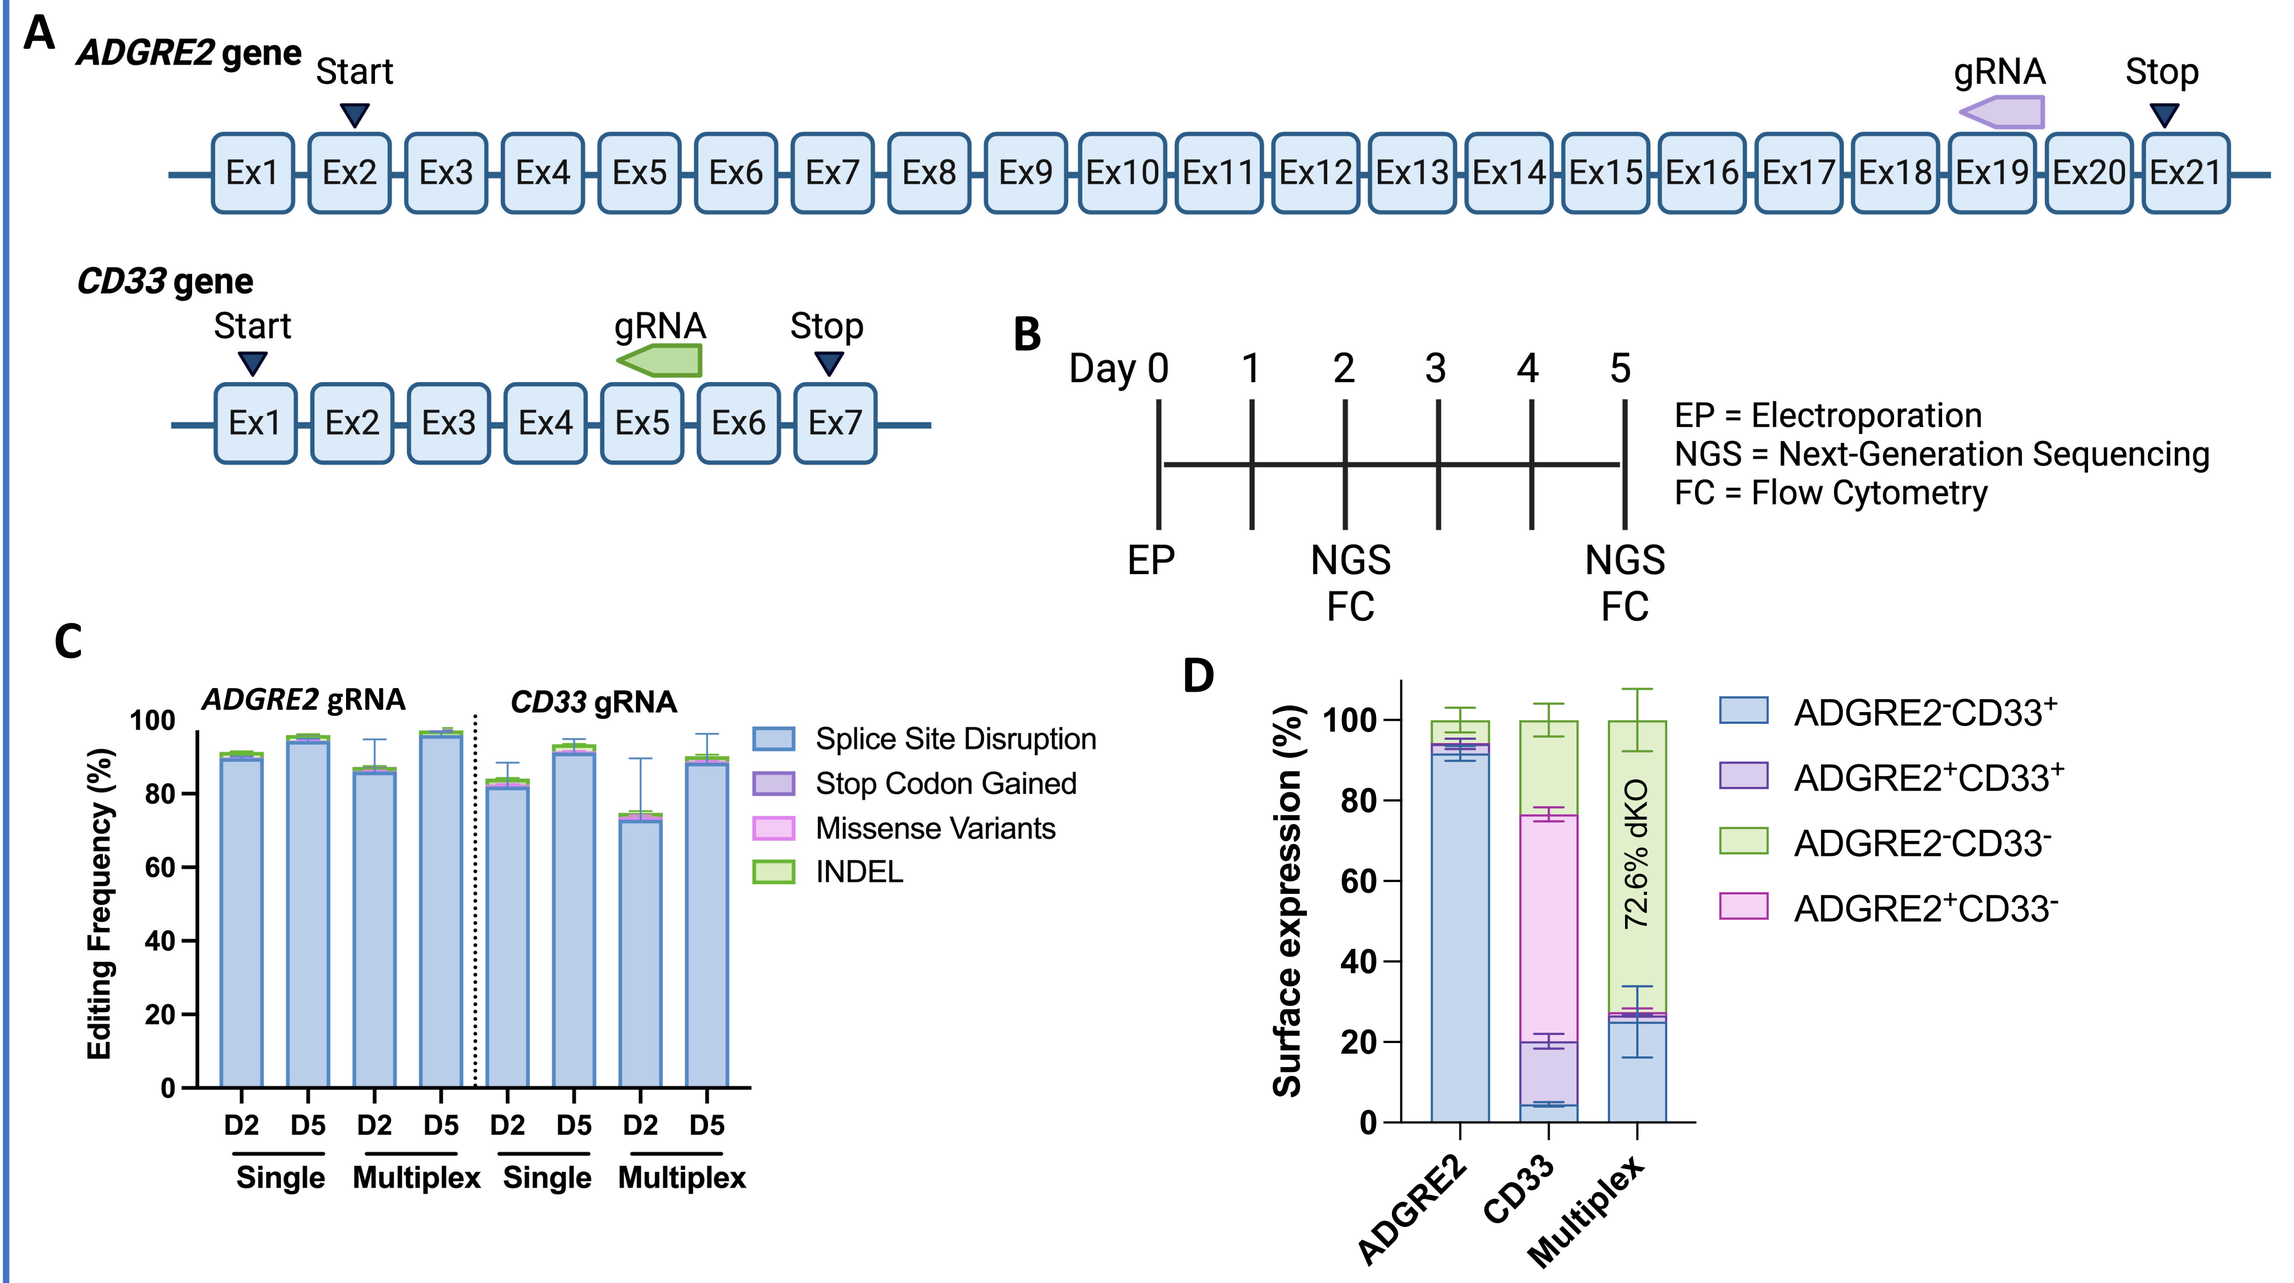
A
ADGRE2 gene
CD33 gene
Start
gRNA
Stop
B
Day 0 1 2 3 4 5
EP
NGS FC
EP = Electroporation
NGS = Next-Generation Sequencing
FC = Flow Cytometry
C
ADGRE2 gRNA
CD33 gRNA
Editing Frequency (%)
Splice Site Disruption
Stop Codon Gained
Missense Variants
INDEL
Single
Multiplex
D
Surface expression (%)
ADGRE2⁻CD33⁺
ADGRE2⁺CD33⁺
ADGRE2⁻CD33⁻
ADGRE2⁺CD33⁻
72.6% dKO

A. Gene structure of *ADGRE2* and *CD33* indicating the adenosine base editing (ABE) gRNA locations. **B.** Experimental schema. **C.** Frequency of editing types at day 2 (D2) and day 5 (D5) after electroporation of HSPCs with ABE guides. **D.** Percentage of the ADGRE2 and CD33 surface protein expression of *ADGRE2* and *CD33* edited cells, in single and multiplex settings, at day 5 (D5). N=2 donors. Data shown as mean ± standard deviation.

CONCLUSIONS

- ADGRE2 is expressed on a high percent of patient AML blasts and LSCs at similar expression intensities at diagnosis and relapse timepoints
- Novel ADGRE2-directed CAR-Ts, engineered with phage display - derived scFv and V_H binders, mediate potent cytolytic activity against ADGRE2-expressing AML cells *in vitro*
- Surface protein expression analysis of ADGRE2 in healthy bone marrow HSPCs and peripheral blood lineages suggests that genetic editing may be necessary to protect healthy hematopoietic lineages from subsequent immunotherapeutic targeting
- The identification and validation of naturally occurring loss-of-function genetic variants of ADGRE2 provides compelling evidence that it is biologically dispensable. Validation studies in endogenous setting are ongoing
- ADGRE2-edited HSPCs demonstrate long-term engraftment, multilineage differentiation, and persistence of editing *in vivo*
- Highly efficient multiplex base editing of ADGRE2 and CD33 leads to robust surface protein KO of both proteins with no impact on myeloid differentiation
- Multiplex genome editing of HSCs paired with subsequent multi-specific immunotherapy can overcome the concerns of tumor heterogeneity and escape mechanisms related to single antigen down-regulation, thereby transforming the current treatment approach for AML

References

1. Zhu X, et al. Nat Struct Mol Biol. 2019;26(8):679-685. 2. Borot F, et al. Proc Natl Acad Sci U S A. 2019;116(24):11978-11987. 3. Humbert O, et al. Sci Transl Med. 2019;11(503):eaaw3768. 4. Kim MY, et al. Cell. 2018;173(6):1439-1453.e19., 5. Lin H, et al. Genomics. 2000; 67(2):188-200. 6. Langenhan T, et al. Sci Signal. 2013; 6(276). 7. Stacey M, et al., Blood 2003;102(8):2916-24 8. I KY, et al. Front Immunol. 2017;8:373.
9. Perna F, et al. Cancer Cell 2017. 32(4): 506-519.e5 10. The Genome Aggregation Database (gnomAD). 11. Boyden S, et al. NEJM 2016; (374)7: 656-663. 12. I KY, et al. Front Immunol. 2020;11:602016.

Certain illustrations created with BioRender.com

Acknowledgments

We thank Jaime Sanchez, Ciara Tucker, Kerry Chios and Lab Operations. Figures 6 and 8 were made with Biorender. We would like to acknowledge Abound for work performed under a collaboration agreement.

Presented at ASCGT 27th Annual Meeting, May 6-10, 2024; Baltimore, MD